#4596 : DUAL-SENSITIZED T-CELLS RESPONDING TO EBV BLCL AND EITHER CMVpp65 OR WT-1 PEPTIDE POOLS HAVE DISTINCT OR SHARED HLA RESTRICTIONS THAT MAY DEPEND ON THE PRESENTING HLA ALLELES..

Ekaterina Doubrovina, Aisha Hasan, Susan Prockop, Karim Baroudy, Richard J. O'Reilly.
Memorial Sloan-Kettering Cancer Center, New York, NY, USA.

ABSTRACT:

Adoptive Immunotherapy with virus-specific T-cells generated from transplant or third party donors can induce durable remissions of severe infections or EBV lymphomas post-transplant. T-cells sensitized with antigens from multiple viruses have also shown promise. However, in any individual donor, immunogenic peptides from different viruses might be expected to elicit T-cell responses restricted by different HLA alleles. In HLA non-identical patients, the efficacy of T-cells reactive against any one virus would be eliminated if the T-cells specific for that virus are restricted by an HLA allele not shared by the patient.

To examine this hypothesis, we evaluated the HLA restrictions of T-cells generated from 42 healthy donors after dual sensitization with either autologous EBV-transformed B-cells (EBVBLCL) loaded with a pool of overlapping 15-mer peptides spanning the sequence of CMVpp65 (n=20) or autologous EBVBLCL loaded with a pool of 15-mers spanning the oncofetal protein WT-1 (n=22). The HLA restrictions of the CMVpp65-specific and WT1 specific T cells were assessed by their cytotoxic activity against a panel of Cr51 labeled dendritic cells sharing a single HLA allele with the T cells donor. The EBV restrictions of the dual sensitized EBV CTLs were identified by their cytotoxic activity against EBV BLCLs sharing the same single HLA alleles derived from the same donors. In 13/20 CMVpp65/EBV sensitized T cells (65%) and 17/22 WT1/EBV sensitized T cells (77%) the CMV or WT1 specific T cell lines were restricted by single HLA alleles. In 10 of the 20 (50%) lines sensitized with EBV BLCL and CMVpp65, CMVpp65 specific T cells were restricted by an HLA allele that was also one of the restricting alleles for EBV CTLs in the same line. However, in the other 10(50%) the CMVpp65 T cells were restricted by an HLA allele different from that of the EBV CTLs. In the 22 lines co-sensitized with EBV and WT1, WT1 specific T cells were restricted by an allele different from those of the EBV CTLs in 13 (59%) lines.

Comparison of EBVCTLs from dual sensitized T cell lines with EBVCTLs contemporaneously generated from the same donors but sensitized with EBV BLCL alone revealed that in 2/4 CMVpp65/EBV lines and 2/5 WT1/EBV lines in which the HLA restriction of CMVpp65 or WT1 specific T cells differed from that of EBV T cells in the same culture, the HLA allele differentially presenting the CMV or WT1 antigen but not an EBV antigen in the dual sensitized cultures was a prominent restricting allele of T cells sensitized with an autologous EBV BLCL alone.

In our bank of 135 CMVpp65-specific T-cells sensitized with autologous APCs loaded with the same pool of overlapping CMVpp65 peptides, T-cells specific for epitopes presented by HLA B0702 were dominant in 33/34 donors inheriting this allele. Furthermore, for T-cell lines generated from 50 donors inheriting HLA A0201, HLA A0201 restricted T-cells specific for the NLV peptide of CMVpp65 were dominant for all lines except those 13 that co-inherited HLA B0702.

Given this dominance of CMVpp65 epitopes presented by HLA-B0702 and HLA-A0201, it is of interest that in the dual sensitized lines, of the 10 in which the HLA restriction of the CMVpp65 specific T-cells differed from that of the EBV-specific T-cells, 5 inherited HLA B0702 or HLA A0201. In each case, the CMVpp65-specific T-cells were restricted by HLA B0702 (N=3) or HLA A0201 (N=2), while the EBV T-cells had a different restriction. However, in T-cells from one of these donors sensitized with EBVBLCL alone, HLA B0702 restricted EBVCTLs were prominent. Furthermore, of the 10 dual-sensitized T-cell lines in which both CMV and EBV specific T-cells restricted by the same allele were detected, the shared restriction was either HLA B0702 (N=2) or HLA A0201 (N=2) in each of the lines inheriting one of these alleles.

Taken together, these results demonstrate that in dual-sensitized T-cells, the HLA restrictions of T-cells specific for one virus or tumor antigen may differ from those of the other in ≥ 50% of cases. While immunodominant epitopes presented by certain prevalent HLA alleles such as HLA B0702 or HLA A0201 often induce responses to both viruses, our findings also suggest that in certain dual-sensitized cultures, immunogenic epitopes from different viruses may compete for presentation by a specific HLA allele or differ in their potential to induce a dominant T-cell response.

BACKGROUND:

- Adoptive immune cell therapy has been proven to be safe and efficient in prophylaxis and treatment of viral complications in immunocompromised patients in post-transplant period
- Antigen-specific CTLs generated in vitro over a period of 3-4 weeks by sensitization with the autologous antigen-presenting cells, such as EBV transformed B cells or dendritic cells presenting the desired array of immunogenic epitopes, are depleted of alloreactive T-cells
- T-cell recognizes virus infected and malignant cells via the specific interaction of its TCR with antigen-derived epitope presented on the restricting HLA allele.
- Expression of at least one HLA allele on the antigen-positive target cells matching the restricting HLA allele of the antigen-specific T-cells is sufficient to trigger CTL induced lysis of the target cells in vitro
- In the in vivo mouse model human EBV specific HLA restricted T-cells, administered intravenously, migrate to the pre-established allogeneic EBV^+ tumor xenograft expressing the restricting HLA allele and induce tumor regression.
- Pre-generated banked virus- and tumor-specific T-cells derived from normal 3rd party donors and pre-characterized for their antigen-specificity and HLA restriction provide an off shelf reagent for immediate treatment of lethal viral complications or malignancies resistant to conventional therapies.
- 3rd party derived virus-specific T-cells can induce durable remissions in patients with severe infections or EBV-lymphomas post-transplant.
- Simultaneous in vitro sensitization of the T-cells of the 3rd party origin with antigens from multiple viruses is an attractive approach
- However, immunogenic epitopes from different viruses may elicit T-cell responses restricted by different HLA alleles not always shared with the patient. Thus, the multi-antigen sensitized T-cells will be deprived of the activity against one or more viruses if they recognize these viruses in the context of the HLA alleles not shared with the patient.

STUDY OBJECTIVES:

1. To characterize the HLA restriction of the CMV specific T-cells and EBV specific T-cells contained within the dual virus specific CTLs simultaneously stimulated with both antigens
2. To characterize the HLA restriction of the WT1 specific T-cells and EBV specific T-cells contained within the CTLs simultaneously stimulated with virus and tumor antigens
3. To compare the patterns of HLA restriction of the EBV specific T-cells generated by in vitro sensitization with unmodified EBV transformed B cells to the EBV specific T-cells developed in the dual CMV/EBV or WT1 /EBV sensitized CTL products.

MATERIALS:

1. PERIPHERAL BLOOD MONONUCLEAR CELLS FROM HEALTHY CONSENTED DONORS
2. 138 PENTADECAPEPTIDES OVERLAPPING BY 11 AMINOACIDS AND SPANNING THE ENTIRE SEQUENCE OF THE CMVpp65
3. WT1 DERIVED 15-MERS GENERATED BASED ON THE 11 AMINOACID OVERLAP AND SPANNING THE ENTIRE SEQUENCE OF THE WT1 PROTEIN
4. MONOCYTE-DERIVED DENDRITIC CELLS, EBV TRANSFORMED B CELL LINES AND PHA ACTIVATED BLASTS

METHODS:

EBV transformed B cell lines (EBV BLCL) were generated in vitro from PBMC of T cell donors by co-incubation with B95-8 EBV concentrate.

Monocyte-derived dendritic cells (DC):
Adherent PBMC were activated in vitro for 7 days in the presence of IL-4, GM-CSF, TNF-α, TGF1β, IL6, PGE2 and tested by FACS analysis for the expression of CD80, CD83, CD86, MHCI and MHC II.

Pools of pentadecapeptides:
WT1 derived pentadecapeptides were mixed into a total pool to provide 0.35mcg of each peptide per ml.
CMVpp65 derived pentadecapeptides were mixed into a total pool to provide 0.17mcg of each peptide per ml.

Cytotoxic T cell lines (CTL):
Generation:
Autologous EBV BLCL were used as stimulators either unmodified or loaded with the total pools of WT1 pentadecapeptides or CMVpp65 pentadecapeptides.
CTLs were cultured in Ysell's medium supplemented with 5% Human Heat Inactivated serum and IL-2 for 3-4 weeks and weekly re-stimulated with chosen antigen-presenting cells.

Characterization of the antigen-specific HLA restriction of CTLs:
HLA restriction of EBV specific T-cells was tested in the ^{51}CR release assay against the panel of allogeneic EBV BLCLs and DCs of the same origin each matching one-two HLA alleles with the CTL donor .
HLA restriction of the WT1 CTLs was determined in the Cr51 release assay against a panel of DCs matching one-two HLA alleles expressed on the donor's T cells either unmodified or loaded with the WT1 peptides.
HLA restriction of the CMV CTLs was determined in the Cr51 release assay against a panel of DCs matching one-two HLA alleles expressed on the donor's T cells either unmodified or loaded with the CMV peptides.

RESULTS:

Representative Examples of HLA restriction of CMV/EBV CTLs and EBV CTLs tested in ^{51}Cr release assay at 25:1 E:T ratio

CMV/EBV DUAL SPECIFIC CTL 061-09

HLA restriction of CMV specific T-cell : B0702 is different than HLA restriction of EBV specific T-cells : A2402

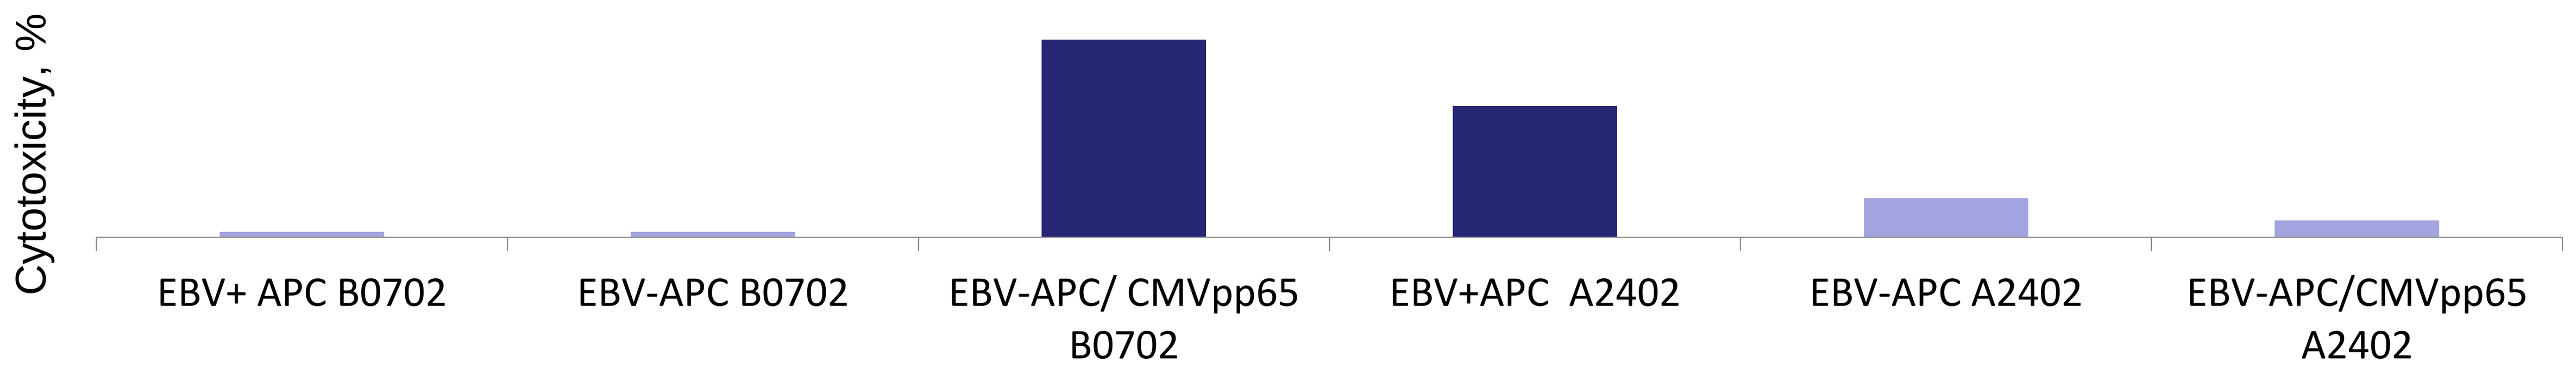
Cytotoxicity, %
EBV+ APC B0702
EBV-APC B0702
EBV-APC/ CMVpp65 B0702
EBV+APC A2402
EBV-APC A2402
EBV-APC/CMVpp65 A2402

CMVpp65 T cells are restricted by an HLA allele different from that of the EBV CTLs

Representative Examples of the patterns of HLA restricting alleles of CMV and EBV specific T-cells within CMV/EBV CTL (n=20)

Donor #	CMV HLA restricting allele	EBV HLA restricting allele
035-11	A01 +1RA	B08
085-12	**A0201**	**A0201** +1RA
061-09	B0702	A2402
026-11	**B0702**	**B0702** +1RA
137-11	**A0201**	**A0201**
110-14	**A2402** +2RA	**A2402**
031-11	B0702	A1101

In 13/20 CMV/EBV CTLs the CMV specific T-cells are restricted by one HLA allele (additional restricting alleles are marked as +1RA or +3RA)

In 10/20 (50%) CMV/EBV dual specific CTL lots CMV-specific T-cells share the Restricting HLA alleles with EBV specific T-cells

Representative Examples of the Comparison of HLA restricting elements of CMV/EBV CTLs and EBV CTLs generated from the same donors (n=4)

	CMV/EBV dual specific T-cells		EBV single specific T-cells
Donor #	CMV HLA restricting allele(s)	EBV HLA restricting allele (s)	EBV HLA restricting allele(s)
061-09	B0702	A2402	A2402 +2RA
031-11	B0702	A1101	B0702 A1101 +3RA

in 2/4 CMV specific T-cells within dual CMV/EBV stimulated CTL products were restricted at least by one same HLA allele as the EBV specific T-cells in the EBV single stimulated CTL products generated from the same donor. In both cases these HLA alleles could not be used as HLA restricting alleles by the EBV specific T-cells in these dual CMV/EBV stimulated CTLs

Representative Examples of the patterns of HLA restricting alleles of WT1 and EBV specific T-cells within WT1/EBV CTL (n=22)

Donor #	WT1 HLA restricting allele	EBV HLA restricting allele
128-11	**B35 +1RA**	**B35 +1RA**
130-11	A0201	A03 +1RA
139-11	A0201	A0101 +4RA
164-11	A1101	B0702 +2RA

In 16/22 WT1/EBV CTLs the WT1 specific T-cells are restricted by one HLA allele

In 7/21 (31%) WT1/EBV dual specific CTL lots WT1-specific T-cells share the Restricting HLA alleles with EBV specific T-cells

Representative Examples of the Comparison of HLA restricting elements of WT1 and EBV specific T-cells in WT1/EBV CTLs and EBV CTLs generated from the same donors (n=6)

	WT1/EBV dual specific T-cells		EBV single specific T-cells
Donor #	WT1 HLA restricting allele(s)	EBV HLA restricting allele (s)	EBV HLA restricting allele(s)
095-09	B35	A32 +1RA	B35 +1RA
139-11	A0201	A0101 +4RA	A0201 +1RA

In 3/5 WT1/EBV CTLs in which the HLA restriction of WT1 specific T cells differed from that of EBV T-cells in the same culture, the HLA allele differentially presenting the WT1 antigen but not an EBV antigen in the dual sensitized cultures was a prominent restricting allele of T cells sensitized with an autologous EBV BLCL alone.

CONCLUSIONS:

1. Simultaneous Sensitization of T-cells with more than one antigen permits generation of the CTLs specific for each of the antigen with distinct HLA restriction
2. in dual-sensitized T-cells, the HLA restrictions of T-cells specific for one of the stimulating antigens may differ from those of the other in ≥ 50% of cases.
3. Certain immunogenic epitopes from different antigens may compete for presentation by a specific HLA allele or differ in their potential to induce a dominant T-cell response if used simultaneously to stimulate same population of T-cells
4. HLA restriction of the multi-antigen specific T-cell products should be defined for each antigen to ascertain the capability of the partially HLA matched CTLs to recognize the antigen of interest in the context of the HLA allele(s) presented on virus-infected or tumor cells of the patient

DISCLOSURE STATEMENT:

Atara Biotherapeutics has an exclusive license to develop and commercialize this cell therapy program.
S.Prockop:
Mesoblast: Research Funding; Atara Biotherapeutics: Research Funding.
E. Doubrovina:
Atara Biotherapeutics: Consultancy, Patents & Royalties, Research Funding.
R.O'Reilly:
Atara Biotherapeutics:Consultancy, Patents & Royalties, Research Funding.