Treatment Patterns for Patients With Post-Transplant Lymphoproliferative Disorder (PTLD) Who Fail Rituximab After Allogeneic Hematopoietic Stem Cell Transplantation: Findings From a Systematic Literature Review

Poster #4777

Hairong Xu,[1] Crystal Watson,[2] Shan Ashton Garib,[3] Anna Forsythe,[3] Arie Barlev[2]

[1]Atara Biotherapeutics, Inc., Westlake Village, CA, USA; [2]Atara Biotherapeutics, Inc, South San Francisco, CA, USA; [3]Purple Squirrel Economics, New York, NY, USA

INTRODUCTION

- Post-transplant lymphoproliferative disorder (PTLD) is one of the most common malignancies following allogeneic hematopoietic stem cell transplantation (HCT).
- In most cases, PTLD is associated with Epstein-Barr virus (EBV) infection.
- The management of PTLD remains a challenge, with no approved treatments for patients.
- Clinical practice treatment guidelines recommend rituximab as first-line therapy for PTLD after allogeneic HCT; however, treatment options for PTLD patients who fail rituximab are not clearly defined.
- We conducted a systematic literature review (SLR) of the published literature to better understand treatment patterns for patients with PTLD who fail rituximab after allogeneic HCT in a real-world setting.

METHODS

Systematic Literature Review

- The SLR was performed following Preferred Reporting Items for Systematic Reviews and Meta-Analyses (PRISMA) guidelines with the scope defined in terms of Population, Intervention Comparators, Outcomes, and Study design (PICOS) criteria.
- Using extensive search terms for the indication and study designs, studies were identified using key biomedical literature databases: EMBASE, MEDLINE, and Cochrane.
- The literature search was conducted on July 19, 2018 and included studies published between database inception in January 1, 1959 and July 19, 2018. Relevant congress abstracts published between January 2015 and June 2018 were also identified.
- The PICOS-based inclusion and exclusion criteria were used to review identified citations.
- No treatment limitations were imposed to ensure inclusion of all relevant evidence; the study designs were limited to prospective and retrospective observational studies. Case reports were included regardless of sample size.
- Two independent reviewers screened all citations and full-text articles; any discrepancies were resolved by a third independent reviewer.
- Data from included studies were extracted into a predefined template, and results were summarized using the PRISMA flow diagram.

RESULTS

PRISMA Flow Diagram

- A total of 447 articles were identified that met the SLR criteria: 350 solid organ transplant (SOT), 70 HCT, and 27 that included both SOT and HCT (**Figure 1**).
- Among the 70 HCT studies, 69 described patients with PTLD after allogeneic HCT, and 48 studies reported data on treatment of patients with PTLD.

Figure 1: PRISMA Flow Diagram

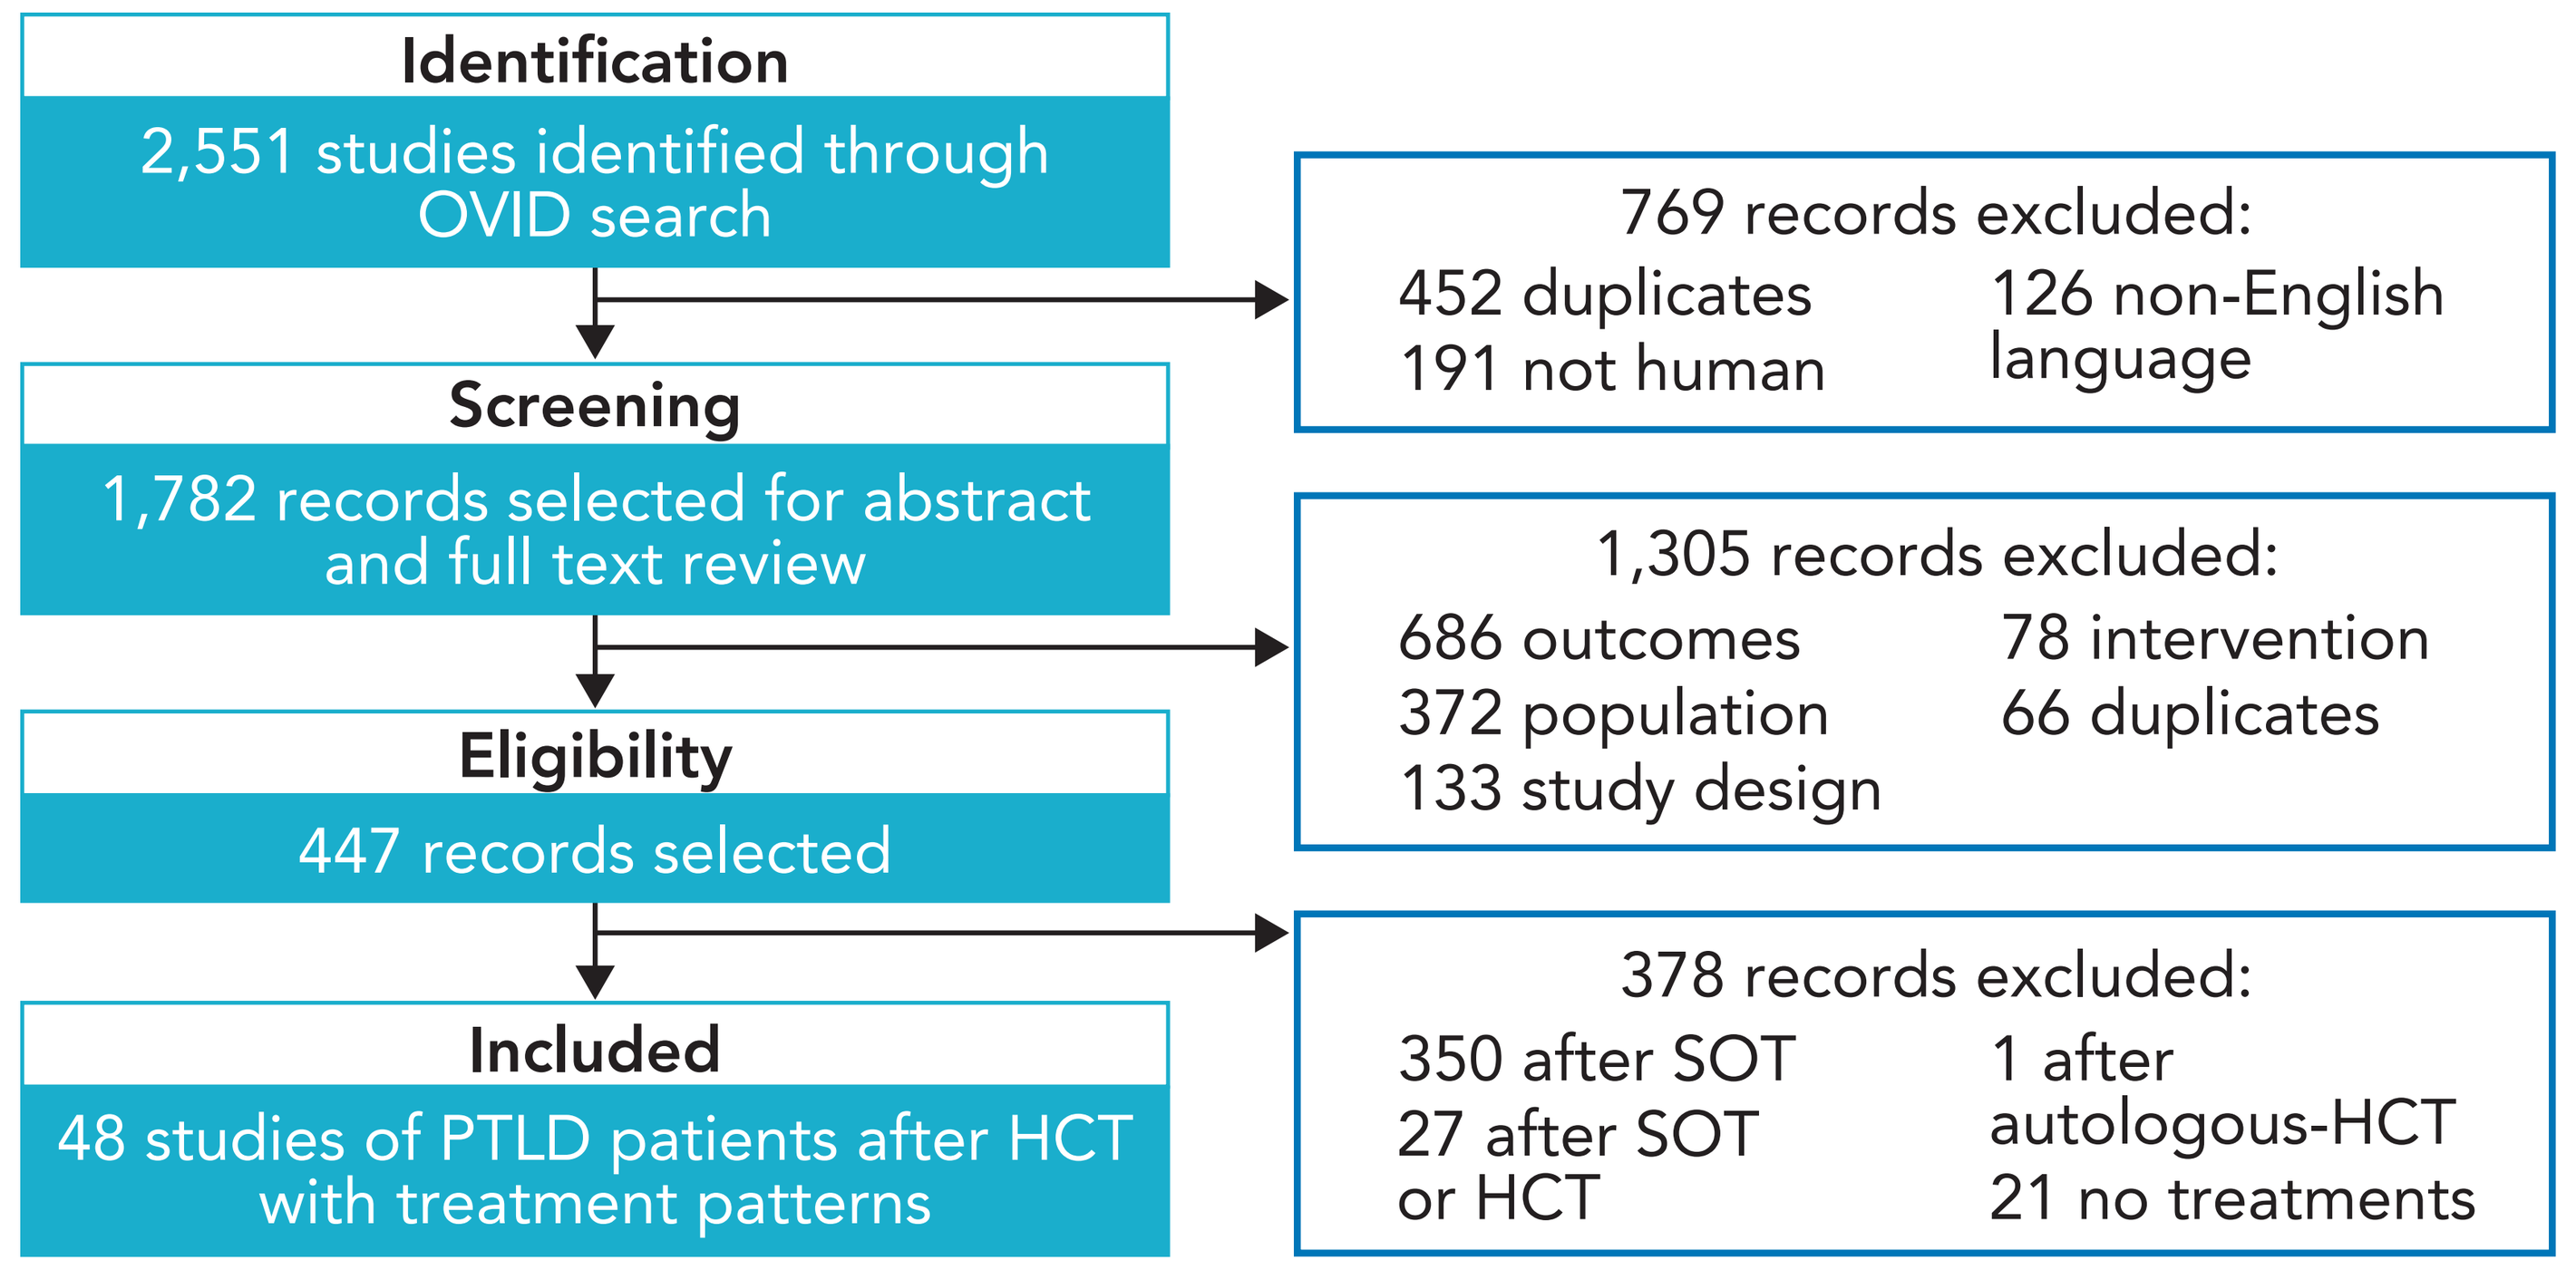
Identification
2,551 studies identified through OVID search
769 records excluded:
452 duplicates
191 not human
126 non-English language
Screening
1,782 records selected for abstract and full text review
1,305 records excluded:
686 outcomes
372 population
133 study design
78 intervention
66 duplicates
Eligibility
447 records selected
378 records excluded:
350 after SOT
27 after SOT or HCT
1 after autologous-HCT
21 no treatments
Included
48 studies of PTLD patients after HCT with treatment patterns

HCT = hematopoietic stem cell transplant; PTLD = post transplant lymphoproliferative disease; SOT = solid organ transplant.

- Most of the included studies were retrospective chart reviews, single-center studies, and reported treatment patterns. Only one study had a sample size of > 100 patients.

Table 1: Description of Studies

Variable	Number of Studies (%)
Age	6 (13%) adult 13 (27%) pediatric 29 (60%) unknown
Study Type	42 (88%) retrospective chart reviews 6 (12%) prospective
Center Type	42 (88%) single-center studies 6 (12%) multi-center studies
Outcomes	46 (96%) clinical outcomes 2 (4%) epidemiology and/or treatments patterns only
Years Covered by Study Period	4 (8%) prior to 2000 34 (71%) 2000–2010 27 (56%) 2010–2016
Sample Size	23 (48%) 1–10 patients 14 (29%) 11–20 patients 4 (8%) 21–30 patients 5 (11%) 40–100 patients 1 (2%) > 100 patients 1 (2%) No sample size reported

First-Line Treatment for PTLD After Allogeneic HCT

- Among studies that reported treatment patterns (total n = 48), first-line therapy in PTLD included rituximab (41 studies), various chemotherapy regimens (15 studies), and lymphocyte infusion (10 studies).

Figure 2: Studies Reporting First-Line Treatments

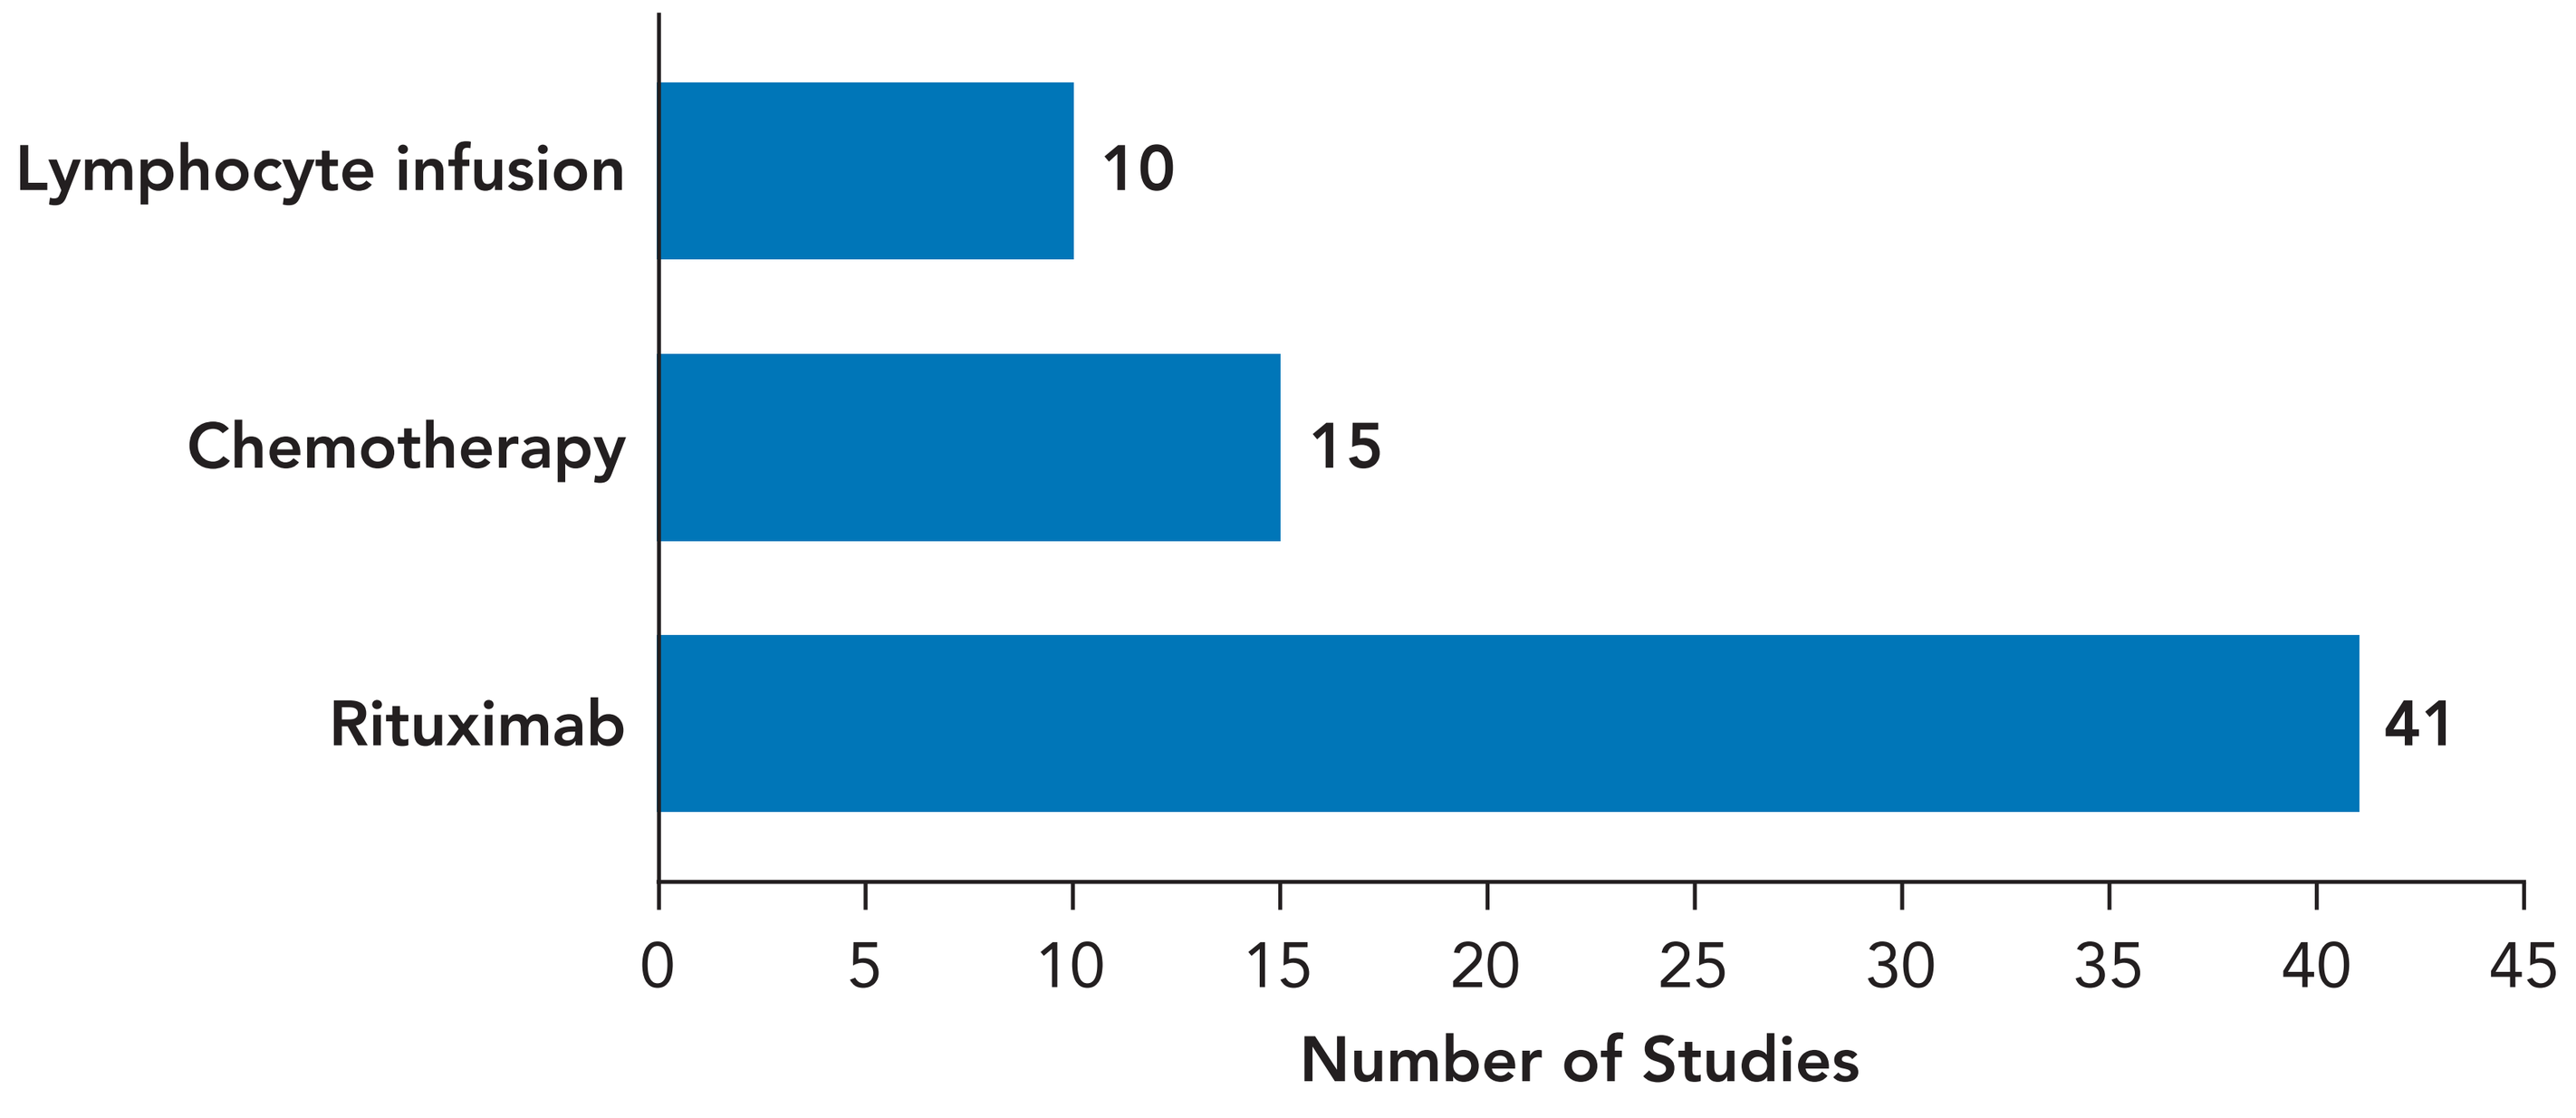
Lymphocyte infusion
10
Chemotherapy
15
Rituximab
41
0
5
10
15
20
25
30
35
40
45
Number of Studies

Second-Line Treatment for PTLD After Allogeneic HCT

- Nine studies reported treatment for patients who failed first-line rituximab (13–67% of PTLD patients); the number of patients with second-line treatments ranged from 2–10 across studies.
- Second-line treatments varied greatly across studies (**Table 2**).
- Only 2 studies (Fox et al, 2014 and Hou et al, 2009[1,2]) reported treatment outcomes in rituximab-refractory patients.

Table 2: Studies Reporting Second-Line Treatment After Rituximab Therapy

Reference	Number of PTLD Patients	2nd Line Treatment	2nd Line Treatment Outcomes
Styczynski et al, 2013[3]	144	Chemotherapy 22%	No outcomes reported
Fox et al, 2014[1]	62	CHOP (2%), rituximab-CHOP (4%), rituximab-CY (2%), rituximab + high-dose cytarabine (2%), lymphocyte infusion (10%)	N = 10; median age = 49 years Chemotherapy: no complete or partial remission Lymphocyte infusion: 60% of patients with complete remission
Poppiti et al, 2016[5]	46	46% second-line treatment (details not reported)	No outcomes reported
Xuan et al, 2013[6]	23	Cytotoxic T lymphocytes (22%), donor lymphocyte infusion (11%)	No outcomes reported
Hou et al, 2009[2]	12	Chemotherapy (25%), donor lymphocyte infusion (33%)	N = 3; mean age = 5 years Lymphocyte infusion: complete remission in 1 patient
Paisiou et al, 2015[8]	15	Chemotherapy (13%)	No outcomes reported
Wu et al, 2012[7]	8	Intrathecal rituximab (60%), donor lymphocyte infusion (% post rituximab not reported)	No outcomes reported
Meyer et al, 2014[9]	5	Rituximab, cyclophosphamide, doxorubicin, vincristine, and prednisone (R-CHOP) + etoposide/cytarabine (VP16/ARA-C), (20%)	No outcomes reported
Hallbook et al, 2014[4]	3	Chemotherapy 67%	No outcomes reported

CONCLUSIONS

- This systematic literature review demonstrates that data on treatment patterns for PTLD patients who failed rituximab after allogeneic HCT are limited (9 studies with a sample size ≤ 10).
- Published data suggest that the percentage of patients who fail rituximab vary greatly (13–67%), there is no consistent standard of care for PTLD patients who fail rituximab, and outcomes are poor.
- There continues to be a significant unmet need among PTLD patients who fail rituximab, and further studies are needed to better understand rituximab response rates in the real-world setting.

REFERENCES: 1. Fox CP, et al. *Bone Marrow Transplant.* 2014;49(2):280–286. 2. Hou HA, et al. *Bone Marrow Transplant.* 2009;43(4):315–21. 3. Styczynski J, et al. *Clin Infect Dis.* 2013;57(6):794–802. 4. Hallbook H, et al. *Bone Marrow Transplant.* Conference: 40th Annual Meeting of the European Group for Blood and Marrow Transplantation. Milan Italy. Conference Publication: (var. pagings). 2014;49(Suppl1):S330. 5. Poppiti K, et al. *Blood.* Conference: 58th Annual Meeting of the American Society of Hematology, ASH 2016. United States. 2016;128(22) (no pagination). 6. Xuan L, et al. *Transplantation.* 2013;96(6)560–566. 7. Wu M, et al. *Blood.* Conference: 54th Annual Meeting of the American Society of Hematology, ASH 2012. Atlanta, GA United States. Conference Publication: 2012:120(21). 8. Paisiou A, et al. *Blood.* 2015;126:5480. 9. Meyer SC, et al. *Hematology.* 2014;19(5):280–5.

ACKNOWLEDGMENTS: We would like to acknowledge Dr. Nazia Rashid for her help with the interpretation of the data and medical writing, and Kathryn Boorer, PhD, for her help with medical writing. **DISCLOSURE:** The study was sponsored by Atara Biotherapeutics, Inc.

Presented at the American Society of Hematology Annual Meeting; San Diego, CA; December 1–4, 2018.