Abstract #4071

Long-Term Outcomes of Subjects with Epstein–Barr Virus-Driven Post-Transplant Lymphoproliferative Disorder (EBV$^+$ PTLD) Following Solid Organ (SOT) or Allogeneic Hematopoietic Cell Transplants (HCT) Treated with Tabelecleucel on a Multicenter Expanded-Access Program

Susan Prockop, MD[1]; Ran Reshef, MD[2]; Donald E. Tsai, MD, PhD[3]; Nancy Bunin, MD[4]; Rolla Abu-Arja, MD[5]; Kris Michael Mahadeo, MD[6]; Wen-Kai Weng, MD, PhD[7]; Koen Van Besien, MD, PhD[8]; David Loeb, MD, PhD[9]; Sunita Dwivedy Nasta, MD[10]; Eneida R. Nemecek, MD, MBA, MSc[11]; Patrick Phuong, PharmD[12]; Minoti Hiremath, MBBS, PhD[12]; Susan Yue, MD[13]; Yan Sun, MS[13]; Willis H. Navarro, MD[12]; Sarah Nikiforow, MD, PhD[14]

[1]Memorial Sloan Kettering Cancer Center, New York, NY; [2]Columbia University Irving Medical Center, New York, NY; [3]Loxo Oncology, South San Francisco, CA; [4]Children's Hospital of Philadelphia, Philadelphia, PA; [5]Nationwide Children's Hospital, Columbus, OH; [6]MD Anderson Cancer Center, Houston, TX; [7]Stanford University, School of Medicine, Stanford, CA; [8]Weill Cornell Medical College, New York, NY; [9]Children´s Hospital at Montefiore, Bronx, NY; [10]University of Pennsylvania, Philadelphia, PA; [11]Pediatric Hematology/Oncology & Bone Marrow Transplantation, OHSU Knight Cancer Institute Doernbecher Children's Hospital, Portland, OR; [12]Atara Biotherapeutics, South San Francisco, CA; [13]Atara Biotherapeutics, Thousand Oaks, CA; [14]Dana-Farber Cancer Institute, Boston, MA

INTRODUCTION

- Patients undergoing solid organ (SOT) or allogeneic hematopoietic cell transplants (HCT) are at risk for developing Epstein–Barr virus-driven post-transplant lymphoproliferative disorder (EBV$^+$ PTLD). Initial treatment of EBV$^+$ PTLD following SOT or HCT usually includes reduction of immunosuppression followed by rituximab ± chemotherapy.
- However, many SOT and HCT recipients are not candidates for chemotherapy due to transplantation, and patients who do not respond to initial treatment have few treatment options and poor outcomes.[1–5] There is an unmet need for effective and well-tolerated therapies in this patient population.
- Tabelecleucel (tab-cel®) is an investigational, off-the-shelf, allogeneic, EBV-specific, T-cell immunotherapy generated from healthy donors that functions through native, endogenous T-cell receptors.
- We have previously shown that treatment with tabelecleucel results in durable responses and a high overall response rate in rituximab-refractory EBV$^+$ PTLD patients (objective response rate [ORR] 68.6% and 50.0% in patients with EBV$^+$ PTLD following HCT and SOT, respectively).[6]
- Here, we report long-term results using tabelecleucel for N=26 patients with EBV$^+$ PTLD following HCT or SOT in EBV-CTL-201 (NCT02822495; active, not recruiting). After December 2018, patients requiring expanded access to tabelecleucel were enrolled in an updated protocol EAP-901 under the same study number. Patients from EAP-901 are not included within the dataset presented here.

METHODS

- EBV-CTL-201 is a multicenter expanded-access protocol to provide access to tabelecleucel for patients with EBV$^+$ diseases for whom there are no appropriate alternative therapies.
- Patients with diverse EBV$^+$ diseases were enrolled on this program, including EBV$^+$ PTLD following HCT or SOT, EBV-associated lymphoproliferative disorders (LPD) associated with primary or acquired immunodeficiency (ID), EBV-associated LPD not associated with ID, EBV$^+$ nasopharyngeal carcinoma, leiomyosarcoma, other EBV$^+$ solid tumors, and EBV viremia.
- Safety analyses are presented for all patients treated with tabelecleucel, regardless of indication. The efficacy analysis focused on outcomes in patients with EBV$^+$ PTLD. An additional efficacy analysis was performed in a subset of EBV$^+$ PTLD patients who had adequate performance status, no central nervous system (CNS) disease, and did not require PTLD-related ventilatory support (consistent with Atara's Phase 3 study population).
- Key inclusion criteria were: presence of EBV$^+$ disease, adequate organ function (absolute neutrophil count ≥500/μL ± cytokine support; platelets ≥20,000/μL ± transfusion support if no grade ≥2 bleed in the preceding 6 months; alanine aminotransferase, aspartate aminotransferase, total bilirubin <3 x upper limit of normal [ULN]; creatinine <3 x ULN), Eastern Cooperative Oncology Group (ECOG) performance status ≤4 or Lansky performance status ≥20, and lack of approved alternative therapies. Non-PTLD-related vasopressor or ventilatory support, ongoing need for methotrexate or extracorporeal photopheresis, and ongoing need for T-cell-inhibiting medications or steroid doses >0.5 mg/kg prednisone were exclusionary. Patients receiving concomitant non-investigational, anti-PTLD therapy were allowed, and are presented in the analysis.
- Patients were treated with 1.6–2.0 x 10^6 cells/kg/dose of tabelecleucel (see **Figure 1**). Tabelecleucel with a different human leukocyte antigen (HLA) restriction was used if patients progressed or were assessed as stable disease for two consecutive cycles; up to four different HLA restrictions were allowed per protocol. Treatment ended with maximal response, occurrence of an adverse event leading to tabelecleucel discontinuation or withdrawal of consent. Maximal response was considered if patients had achieved three consecutive partial response or two consecutive complete response assessments (per Lugano Response Criteria).
- Tabelecleucel manufacturing process involves collecting normal, healthy volunteer donor peripheral blood mononuclear cells, and separating the B and T cells. B cells are then transformed to generate antigen-presenting cells and co-cultured with T cells, along with cytokine stimulation. After EBV cytotoxic T-lymphocyte expansion, the product is characterized and stored for off-the-shelf use.

Figure 1. EBV-CTL-201 Study Schema

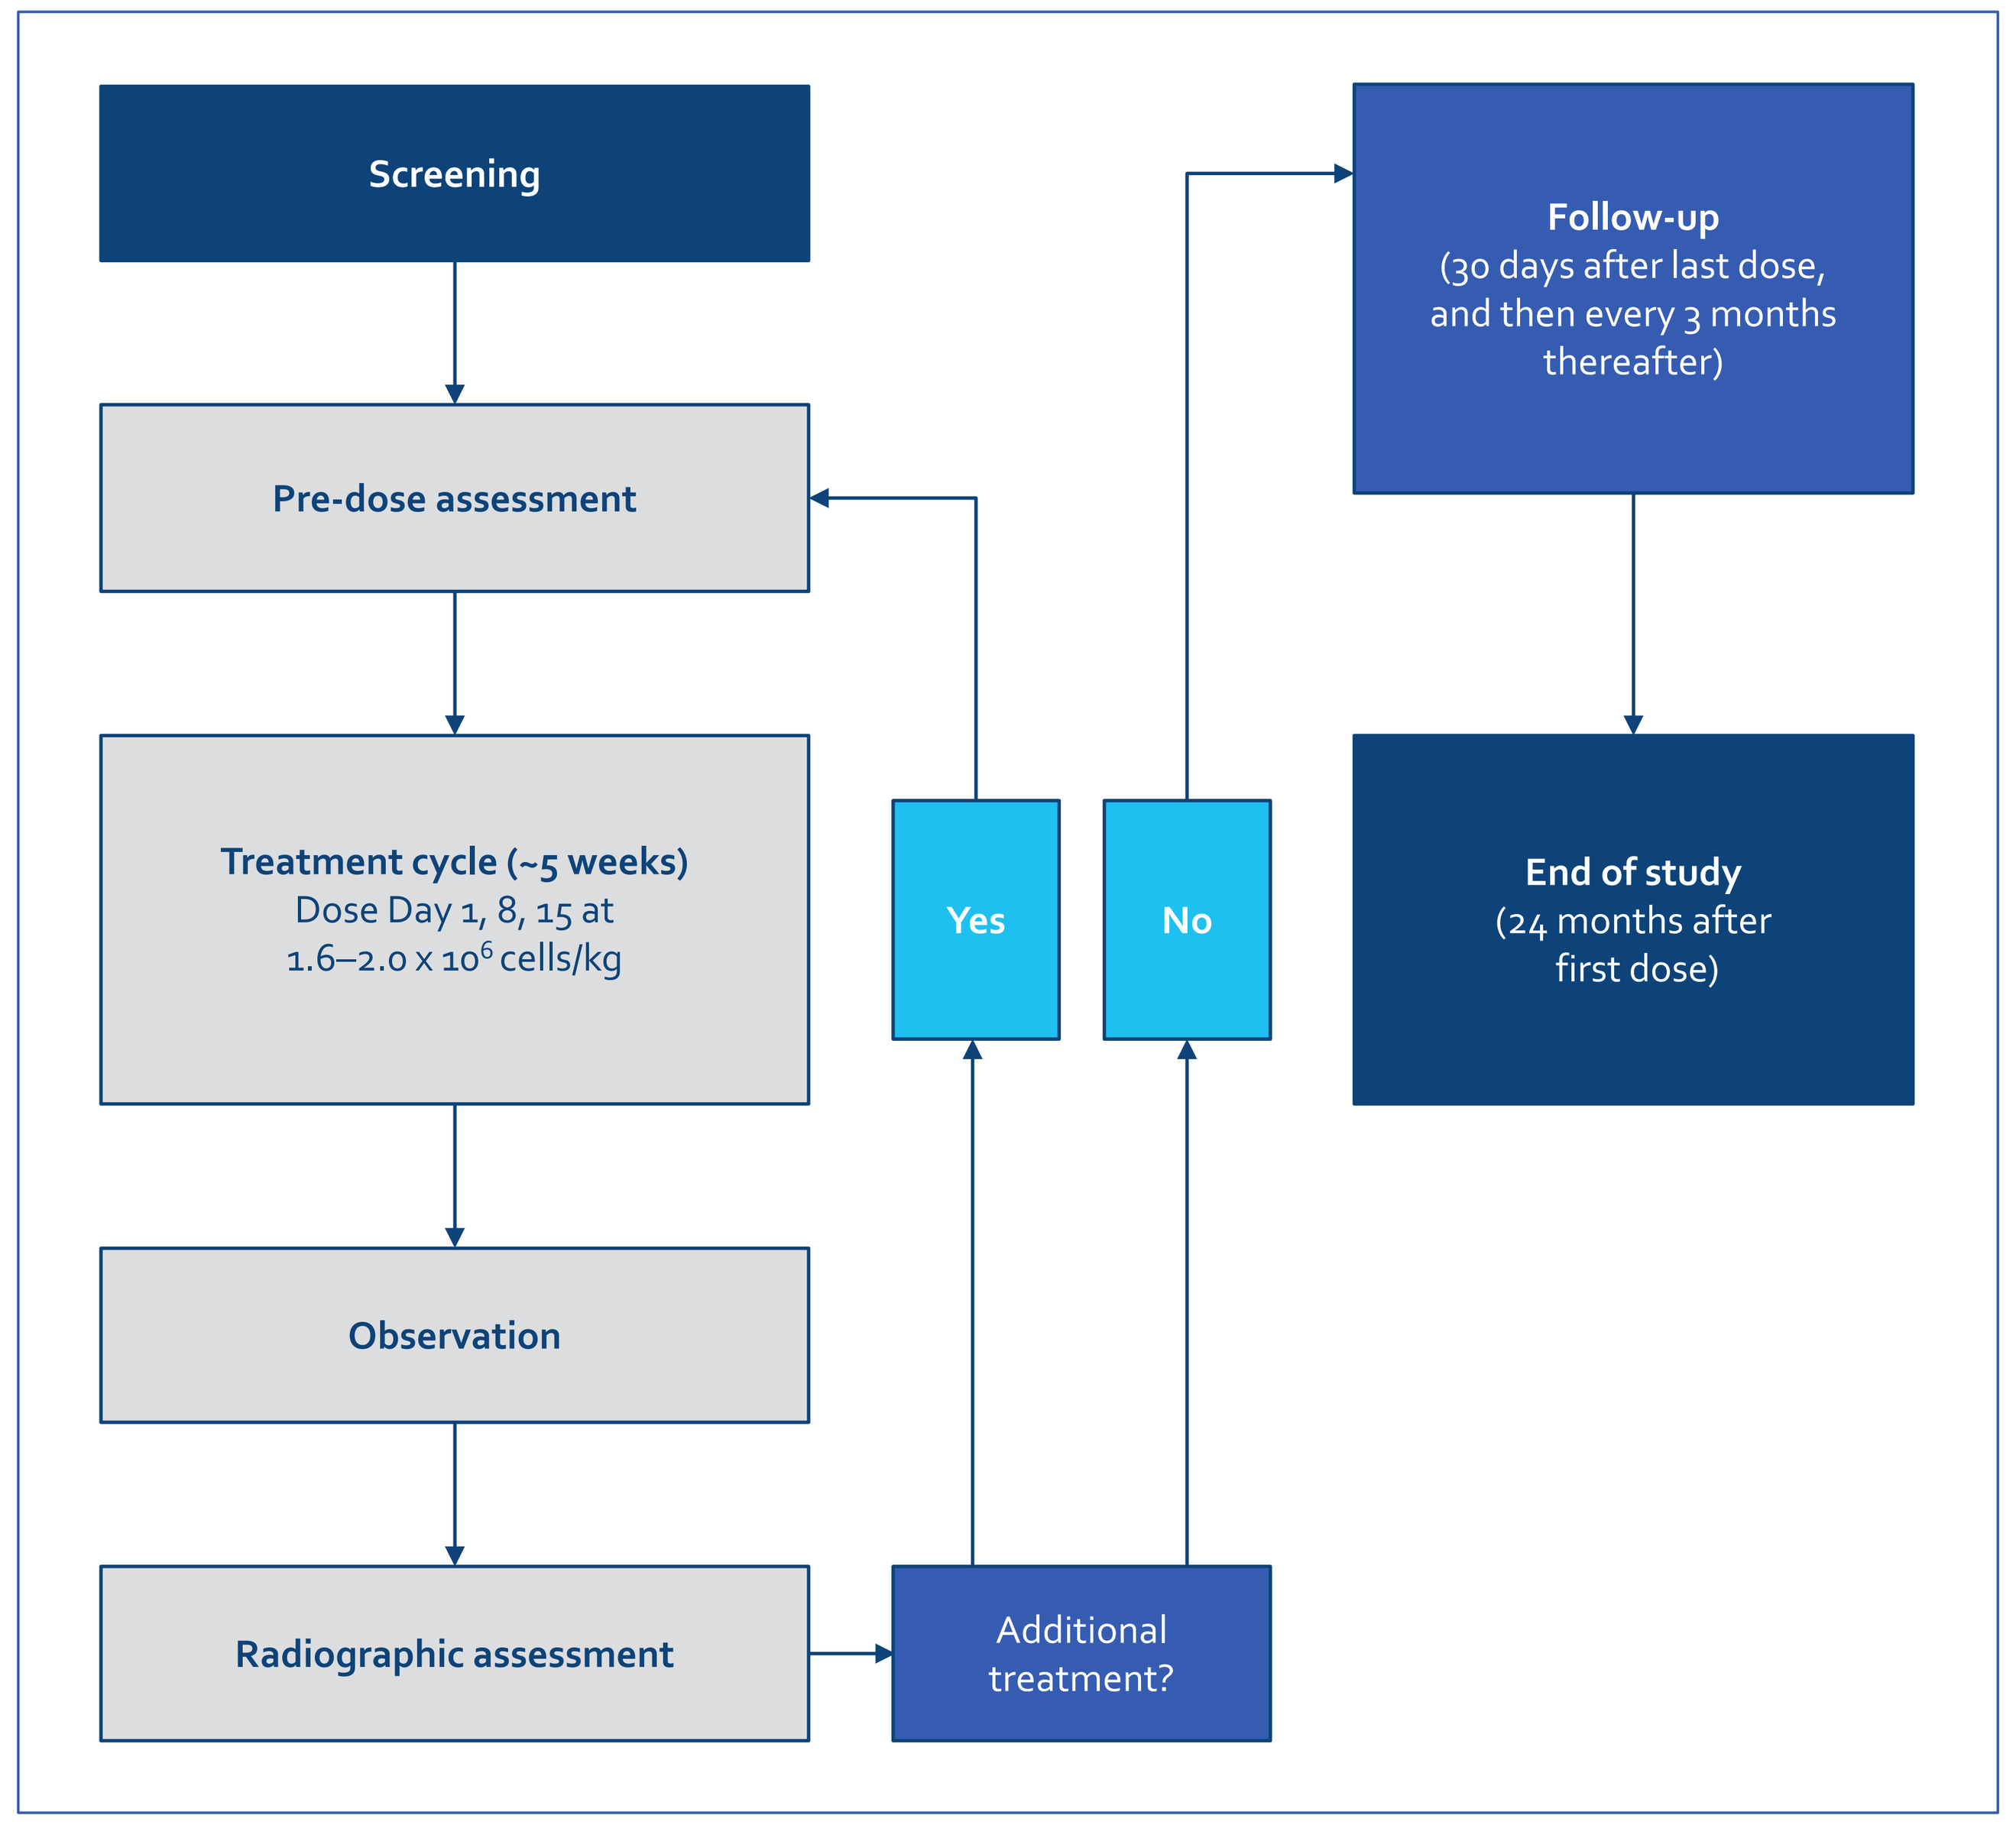

Screening
Pre-dose assessment
Treatment cycle (~5 weeks)
Dose Day 1, 8, 15 at
1.6–2.0 x 10^6 cells/kg
Observation
Radiographic assessment
Additional treatment?
Yes
No
Follow-up
(30 days after last dose, and then every 3 months thereafter)
End of study
(24 months after first dose)

Tabelecleucel was administered on Days 1, 8, and 15 of every 5-week cycle with imaging-based response assessment at ~Day 28 of each cycle.

ENROLLMENT AND DEMOGRAPHICS

- As of September 24, 2019, 26 patients who developed EBV$^+$ PTLD following HCT (n=14) or SOT (n=12) after failure of rituximab ± chemotherapy have been enrolled and received treatment with tabelecleucel. Patient characteristics are shown in **Table 1**.
- Most patients received prior treatment with rituximab monotherapy. Additionally, most patients in the SOT cohort also received prior chemotherapy with regimens, including anthracycline-, platinum-, and/or gemcitabine-based therapy.
- Across both HCT and SOT cohorts, time from transplant to diagnosis of EBV$^+$ PTLD was 5.1 months (range: 1–276) and time from initial EBV$^+$ PTLD diagnosis to first administration of tabelecleucel was 2.3 months (range: 0.2–68).
- According to the PTLD prognostic index,[7] 92% and 63% of patients in the HCT and SOT cohorts, respectively, were considered intermediate/high risk based on one or more of the following: age ≥60 years, ECOG performance status ≥2, and/or elevated baseline lactate dehydrogenase (see **Table 1**).
- Of note, most patients in the HCT (11/14; 79%) and SOT (11/12; 92%) cohorts may have been eligible for Atara's ongoing Phase 3 trials based on adequate ECOG performance status, no CNS disease, and no PTLD-related ventilatory support.
- EBV$^+$ PTLD patients received tabelecleucel at a median dosage of 1.98 x 10^6 cells/kg/dose (range: 1.6–2.0), with a median of 6 doses administered (range: 1–27).

Table 1. Baseline Characteristics – EBV$^+$ PTLD Patients Following Allogeneic Hematopoietic Cell (HCT) and Solid Organ (SOT) Transplant

Characteristics	HCT (n=14)	SOT (n=12)
Age, median (range)	46 (2–74)	27.5 (7–66)
Male, n (%)	7 (50)	6 (50)
Race, n (%)		
Caucasian	10 (71)	8 (67)
Black	1 (7)	0
Asian	2 (14)	1 (8)
Other/unknown	1 (7)	3 (25)
Solid organ transplanted, n (%)		
Kidney	NA	6 (50)
Intestine	NA	1 (8)
Heart	NA	2 (17)
Lung	NA	2 (17)
Other	NA	1 (8)
ECOG PS ≥2 or Lansky PS ≤60, n (%)	8 (57)	4 (33)
Intermediate/high-risk PTLD prognostic index* (%)	92	63
Prior rituximab monotherapy, n (%)	14 (100)	11 (92)
Prior chemotherapy, n (%)	1 (7)	7 (58)

*Choquet S, et al. 2017; in subjects aged ≥16 years (12 HCT and 8 SOT). Intermediate/high-risk patients had one or more of the following: age ≥60 years, ECOG PS ≥2, and/or elevated baseline lactate dehydrogenase.
ECOG = Eastern Cooperative Oncology Group; NA = not available; PS = performance status.

EFFICACY

- A high proportion of EBV$^+$ PTLD patients responded to tabelecleucel, with median time to response of 1 month (range: 0.6–7). Of note, ORR was higher in the SOT versus HCT cohort, which may be reflective of higher rates of intermediate/high-risk patients in the HCT cohort (**Tables 2 and 3**).
- Patients responding to tabelecleucel had favorable overall survival (OS) compared with non-responders (**Figures 2 and 3**). Among EBV$^+$ PTLD patients responding to tabelecleucel, no deaths attributable to PTLD progression have been reported.
- Median follow-up time was 3 months (range: 1–25) and 18 months (range: 3–26) in HCT and SOT cohorts, respectively. Though median follow-up in HCT patients was short, 4 patients (29%) were followed up for >12 months.

Table 2. Efficacy for Patients With EBV$^+$ PTLD – Allogeneic Hematopoietic Cell Transplant (HCT) Cohort

HCT Cohort	All HCT (n=14)	Potential Ph3 Subset* (n=11)	Responders (n=7)
ORR (investigator assessed), n (%)	7 (50)	6 (55)	–
1-year OS, % (95% CI)	60 (29, 81)	79 (38, 94)	86 (33, 98)
2-year OS, % (95% CI)	60 (29, 81)	79 (38, 94)	86 (33, 98)

*Based on adequate ECOG performance status, no CNS disease, and no PTLD-related ventilatory support.
CI = confidence interval; ECOG = Eastern Cooperative Oncology Group; ORR = objective response rate; OS = overall survival; Ph3 = Phase 3.

Table 3. Efficacy for Patients With EBV$^+$ PTLD – Solid Organ Transplant (SOT) Cohort

SOT Cohort	All SOT (n=12)	Potential Ph3 Subset* (n=11)	Responders (n=10)
ORR (investigator-assessed), n (%)	10 (83)	9 (82)	-
1-year OS, % (95% CI)	83 (46, 95)	81 (42, 95)	100
2-year OS, % (95% CI)	83 (46, 95)	81 (42, 95)	100

*Based on adequate ECOG performance status, no CNS disease, and no PTLD-related ventilatory support.
CI = confidence interval; ECOG = Eastern Cooperative Oncology Group; ORR = objective response rate; OS = overall survival; Ph3 = Phase 3.

Figure 2. Kaplan–Meier Plot of Overall Survival by Response in Patients With EBV$^+$ PTLD – Hematopoietic Cell Transplant Cohort

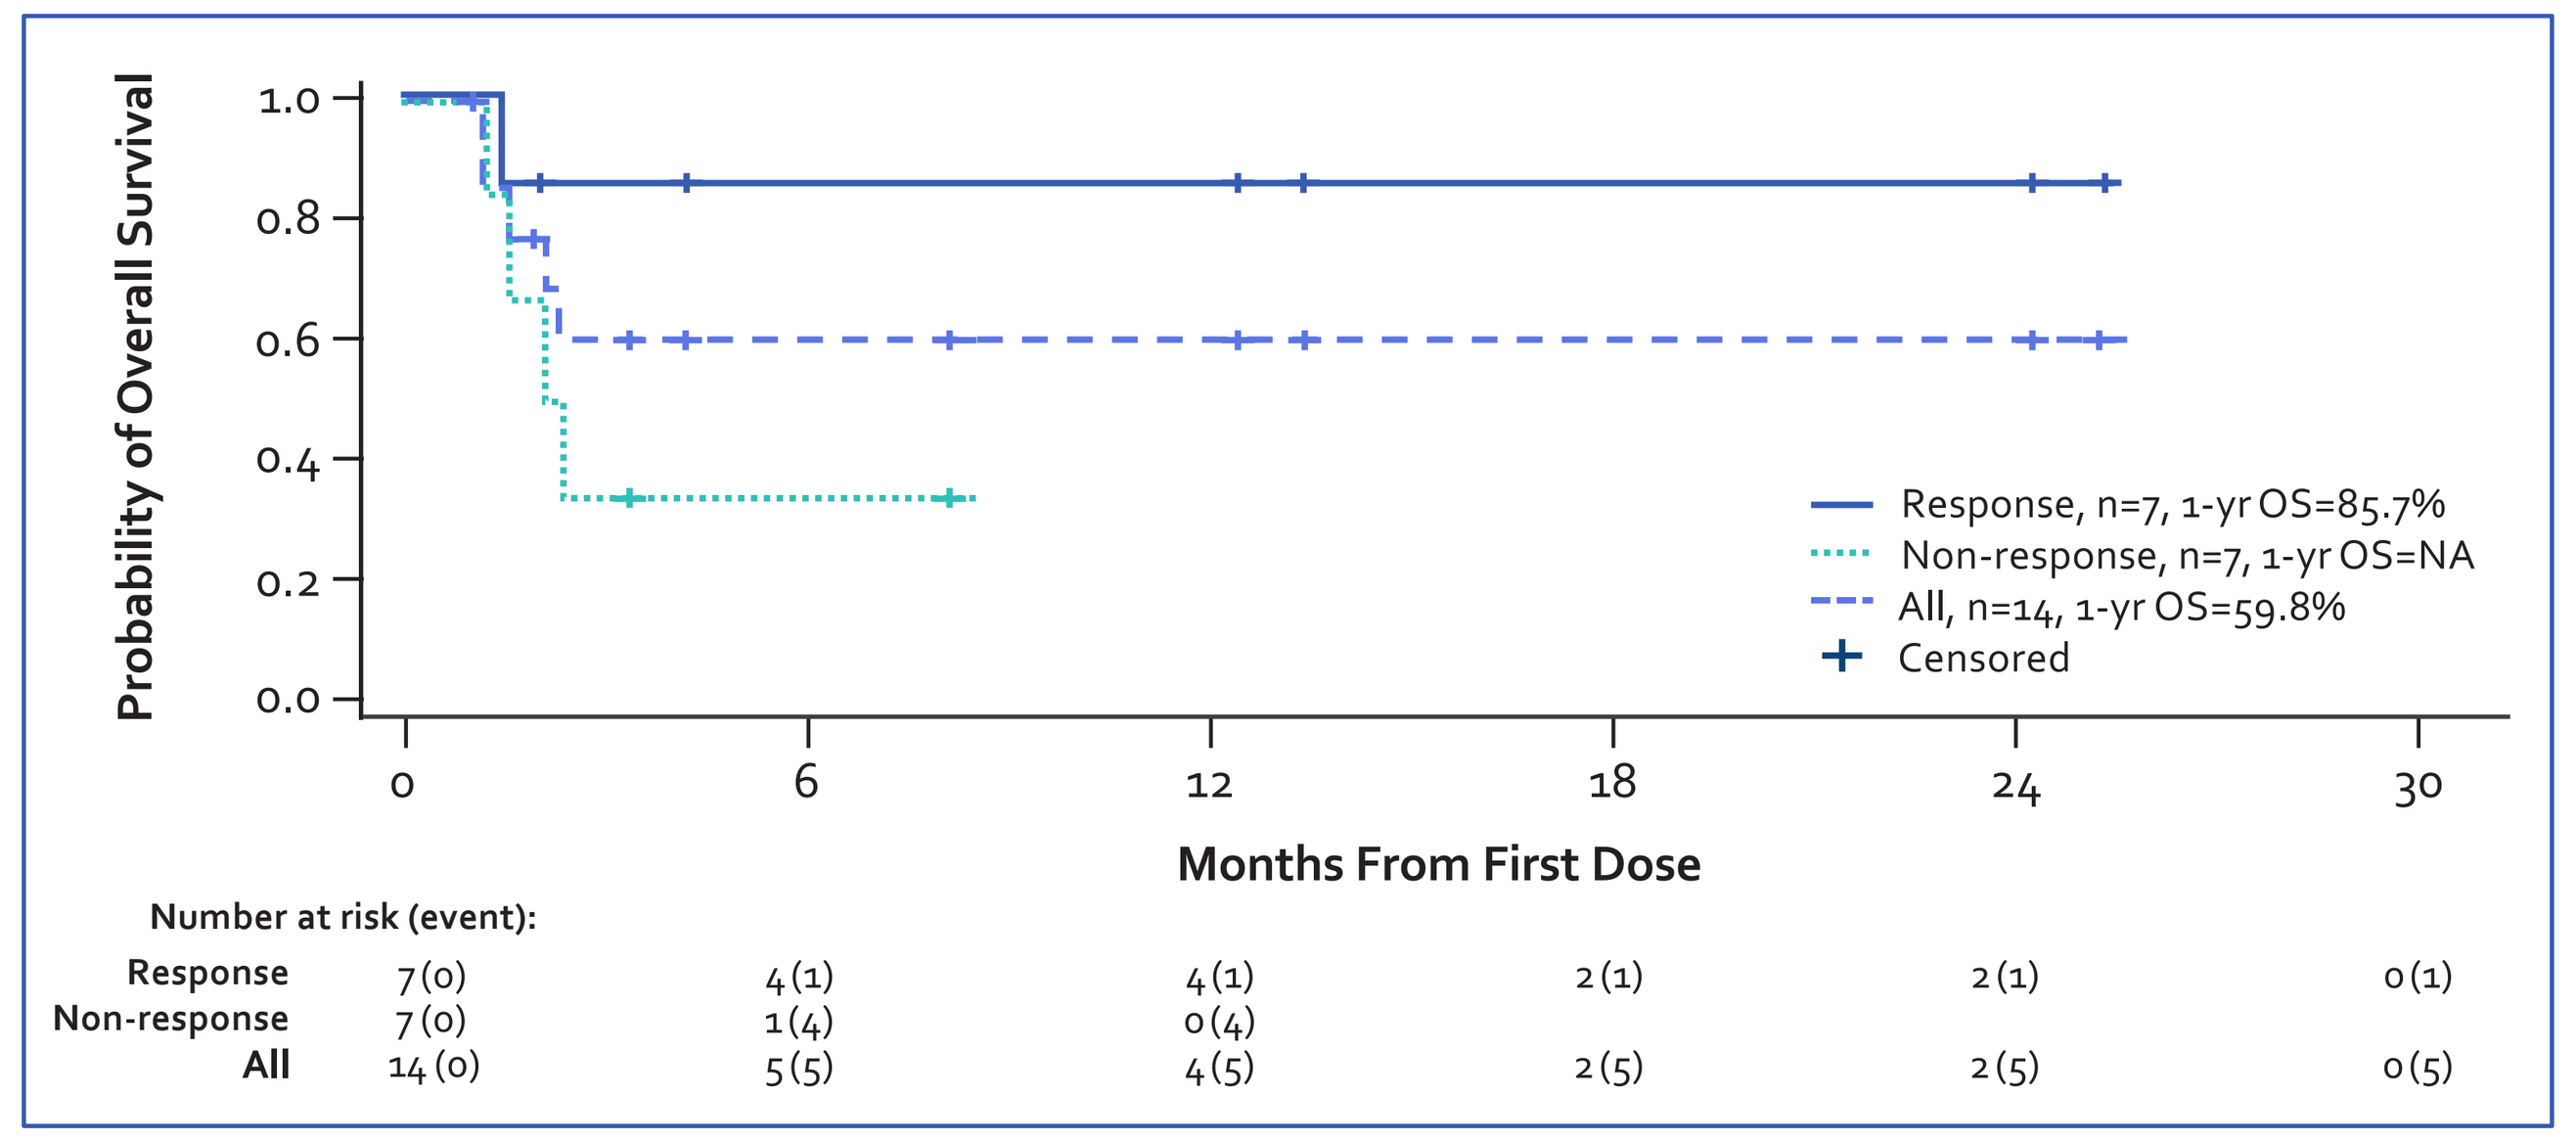

Probability of Overall Survival
1.0
0.8
0.6
0.4
0.2
0.0
0
6
12
18
24
30
Months From First Dose
Response, n=7, 1-yr OS=85.7%
Non-response, n=7, 1-yr OS=NA
All, n=14, 1-yr OS=59.8%
Censored
Number at risk (event):
Response 7 (0) 4 (1) 4 (1) 2 (1) 2 (1) 0 (1)
Non-response 7 (0) 1 (4) 0 (4)
All 14 (0) 5 (5) 4 (5) 2 (5) 2 (5) 0 (5)

Figure 3. Kaplan–Meier Plot of Overall Survival by Response in Patients With EBV$^+$ PTLD – Solid Organ Transplant Cohort

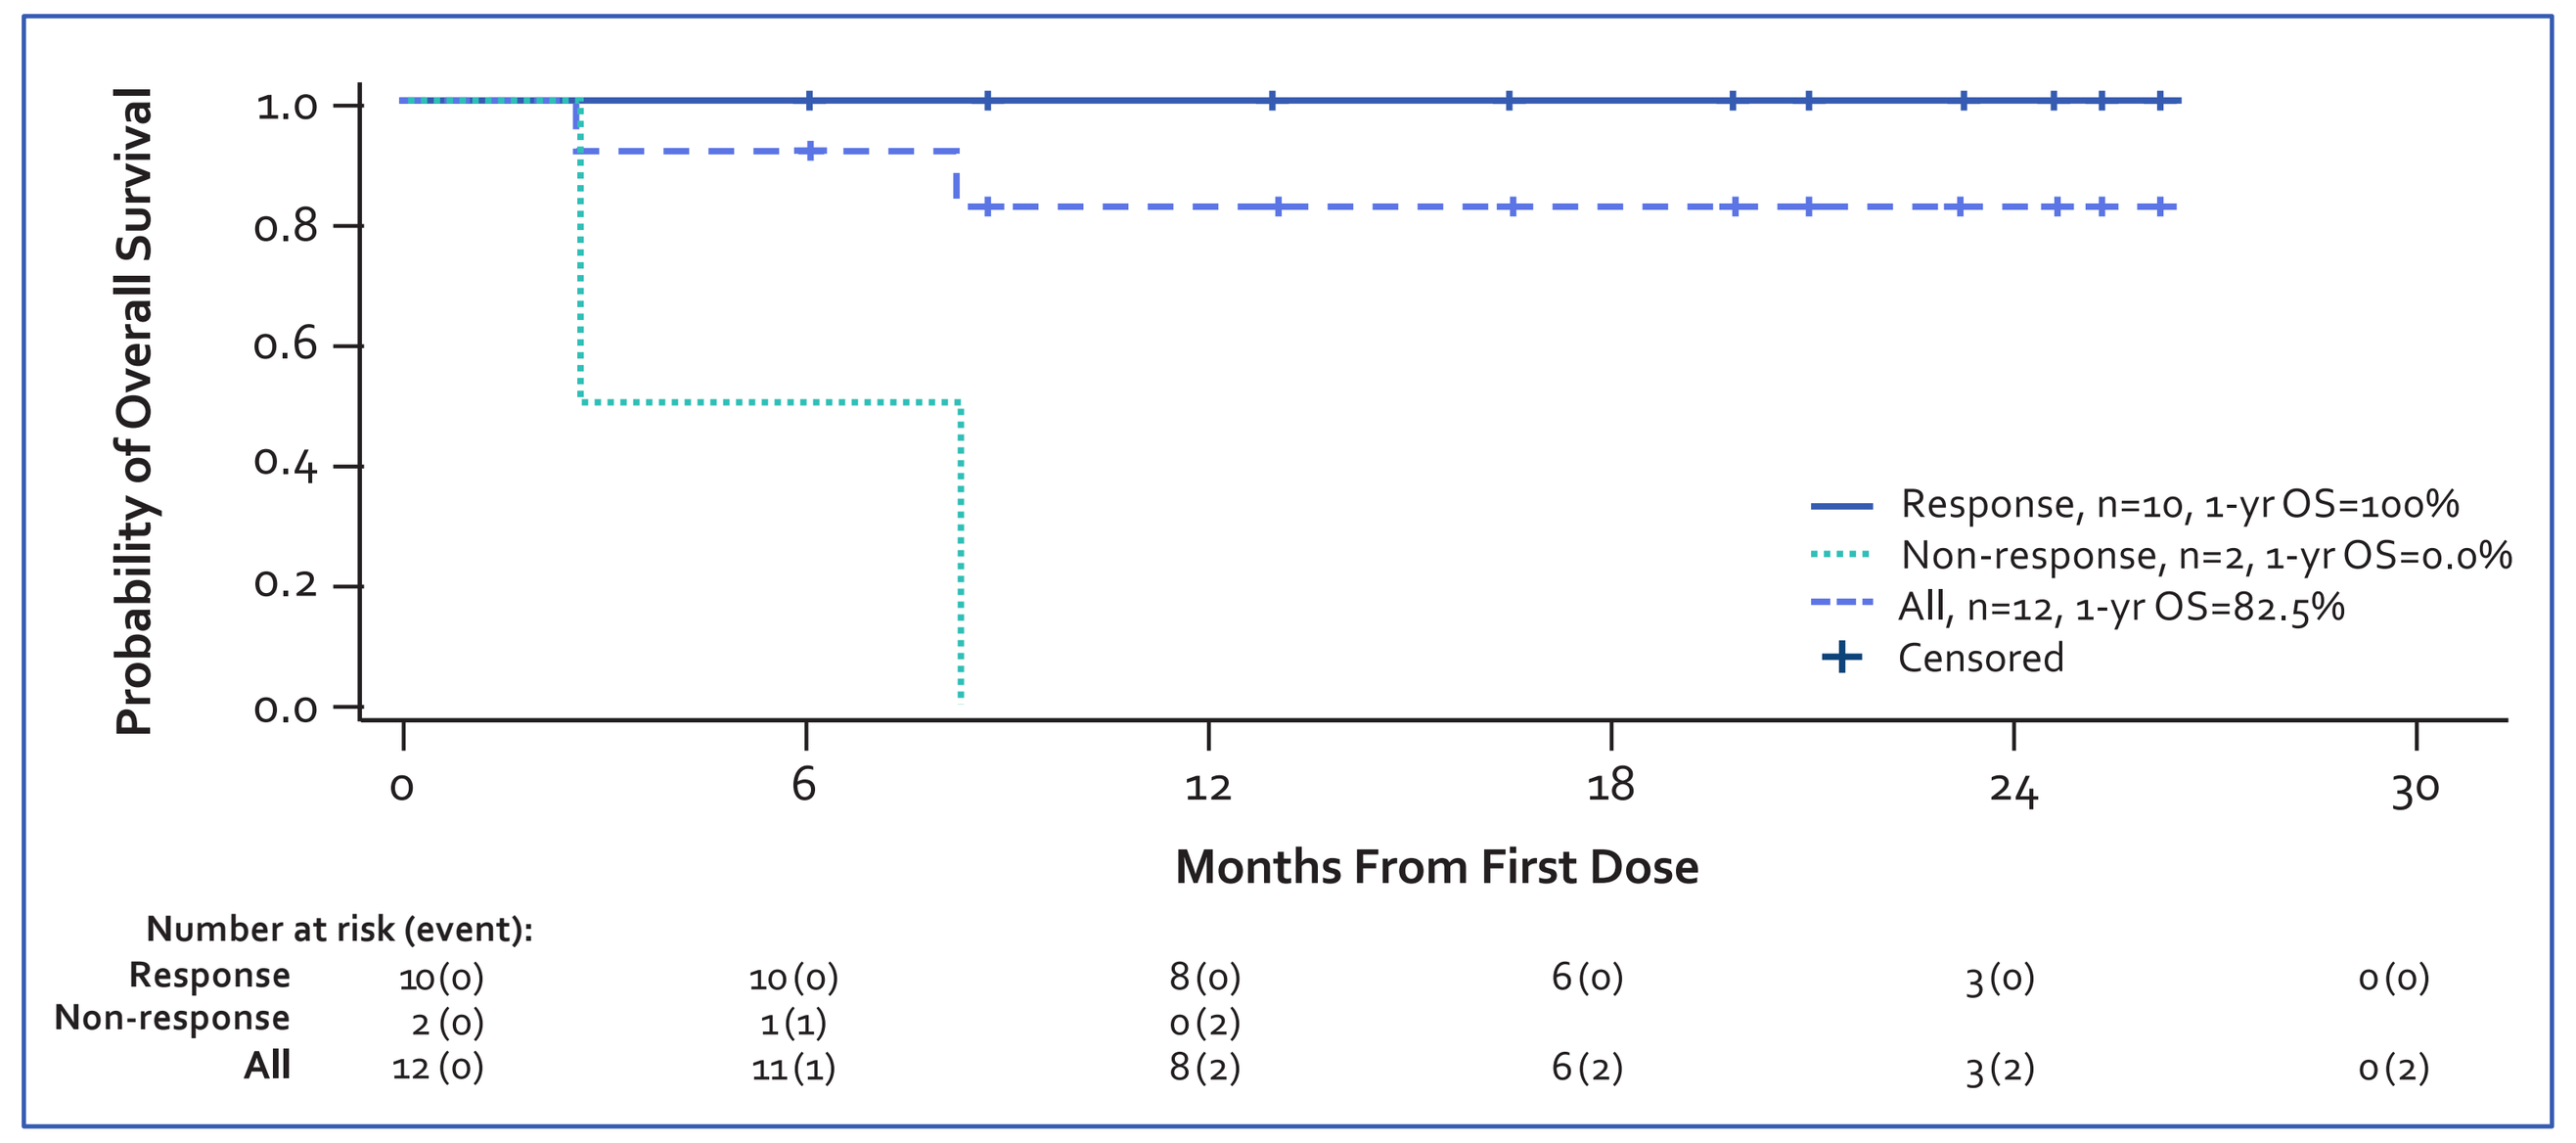

Probability of Overall Survival
1.0
0.8
0.6
0.4
0.2
0.0
0
6
12
18
24
30
Months From First Dose
Response, n=10, 1-yr OS=100%
Non-response, n=2, 1-yr OS=0.0%
All, n=12, 1-yr OS=82.5%
Censored
Number at risk (event):
Response 10 (0) 10 (0) 8 (0) 6 (0) 3 (0) 0 (0)
Non-response 2 (0) 1 (1) 0 (2)
All 12 (0) 11 (1) 8 (2) 6 (2) 3 (2) 0 (2)

Response: patients who achieved objective response of CR or PR; Non-response, patients who failed to achieve objective response of CR or PR.
CR = complete response; PR = partial response; OS = overall survival; NA = follow-up not available.

SAFETY

- The safety analysis population consists of all patients treated in EBV-CTL-201 (N=61), which includes n=26 EBV$^+$ PTLD patients and n=35 patients with other EBV-associated diseases. The most common treatment-emergent adverse events (TEAEs) reported in ≥20% of all patients are shown in **Table 4**. The most common treatment-emergent serious adverse events (TESAEs) reported in ≥5% of all patients were disease progression (16%), pyrexia (8%), and pneumonia (7%). Disease progression was the only fatal TESAE reported in >1 pt (5 patients; 8%).
- Adverse events of special interest of graft-versus-host disease (GvHD) were reported in three patients, all with prior allogeneic HCT. No events of GvHD led to study drug discontinuation. No other adverse events of special interest (cytokine-release syndrome, infusion-related reaction, or transmission of infectious disease) were reported.
- TEAEs and TESAEs that occurred from start of tabelecleucel to 30 days after the last dose are shown in **Tables 4 and 5**.

Table 4. Most Common TEAEs Reported in ≥20% of All Patients

Preferred Term	All Patients (N=61) n (%)	EBV$^+$ PTLD Patients Only (N=26) n (%)
Pyrexia	20 (33)	9 (35)
Cough	16 (26)	8 (31)
Fatigue	16 (26)	8 (31)
Diarrhea	15 (25)	9 (35)
Aspartate aminotransferase increased	13 (21)	8 (31)
Disease progression	13 (21)	6 (23)

EBV = Epstein–Barr virus; PTLD = post-transplant lymphoproliferative disorder; TEAE = treatment-emergent adverse event.

Table 5. Summary of Safety Profile of Tabelecleucel in All Patients

Event Type	All Patients (N=61) n (%)
All TEAEs	**60 (98)**
Grade ≥3 TEAEs	39 (64)
All treatment-related TEAEs	**26 (43)**
Grade ≥3 treatment-related TEAEs	9 (15)
All TESAEs (all grades)	**33 (54)**
Treatment-related TESAEs (all grades)	**7 (12)**
Fatal TESAEs	**9 (15)**

TEAE = treatment-emergent adverse event; TESAE = treatment-emergent serious adverse event.

CONCLUSIONS

- **Results from this analysis demonstrate a high ORR, short time to response, and favorable estimated long-term OS for tabelecleucel in patients with EBV$^+$ PTLD following HCT or SOT who have high-risk clinical features and previously failed rituximab ± chemotherapy.**
- **Similarly encouraging outcomes were observed in the subset of EBV$^+$ PTLD patients who would have been potentially eligible for ongoing Phase 3 studies of tabelecleucel.**
- **Tabelecleucel was generally well-tolerated in a population of mostly immunocompromised EBV$^+$ PTLD patients with multiple comorbidities. The tolerable safety profile of tabelecleucel was consistent in patients treated with EBV-associated diseases.**
- **Tabelecleucel has been shown to be an effective treatment option with a compelling benefit:risk profile for patients with EBV$^+$ PTLD following HCT or SOT, who have no approved therapies. These results warrant further clinical investigation and confirmation.**

REFERENCES

1. Zimmermann H *et al. EHA Library* 2019;266518;PF719.
2. Fox CP *et al. Bone Marrow Transplant* 2014;49:280–6.
3. Ocheni S *et al. Bone Marrow Transplant* 2008;42:181–6.
4. Uhlin M *et al. Haematologica* 2014;99:346–52.
5. Choquet S *et al. Haematologica* 2007;92:273–4.
6. Prockop S *et al. EHA Library* 2018;214869;PF401, plus associated poster presentation.
7. Choquet S *et al. Ann Hematol* 2007;86:599–607.

DISCLOSURES AND ACKNOWLEDGMENTS

This study was funded by Atara Bio. Medical writing assistance was provided by Anna Sanniti, PhD from Seques (AMICULUM Ltd), funded by Atara Bio.

Poster PDF

ASH PosterCast