A Multicenter, Open Label, Phase 3 Study of Tabelecleucel for Solid Organ or Allogeneic Hematopoietic Cell Transplant Participants with Epstein-Barr Virus-Driven Post-Transplant Lymphoproliferative Disease (EBV+ PTLD) after Failure of Rituximab or Rituximab and Chemotherapy (ALLELE)

Poster 413

Susan E Prockop, MD,[1] Minoti Hiremath, MBBS, PhD,[2] Wei Ye, PhD,[2] Laurence Gamelin, PhD,[2] Willis H. Navarro, MD,[2] and Kris Michael Mahadeo, MD[3]

[1]Memorial Sloan Kettering Cancer Center, New York, NY; [2]Atara Biotherapeutics, South San Francisco, CA; [3]MD Anderson Cancer Center, Houston, TX

BACKGROUND

EBV+ PTLD

- Patients undergoing solid organ transplant (SOT) or allogeneic hematopoietic cell transplant (HCT) are at risk of developing Epstein-Barr virus-driven post-transplant lymphoproliferative disease (EBV+ PTLD).[1–3]
 - EBV+ PTLD is a rare, aggressive, and potentially deadly hematologic malignancy that occurs following transplantation when T cell activity is compromised by immunosuppression.[1,3]
- Treatments for EBV+ PTLD following SOT and HCT include:
 - Reduction in immunosuppression.[4]
 - Off-label rituximab ± multi-agent chemotherapy.[3,5–8]
- Following failure of rituximab therapy (HCT) or rituximab ± chemotherapy (SOT), current treatments are limited by non-response or low response, relapse, potential for graft rejection, short- and long-term toxicity, and high mortality.[4–8]
 - After failure of rituximab in EBV+ PTLD following HCT, patients died at a median of 33 days from diagnosis.[5]
 - After failure of rituximab ± chemotherapy in EBV+ PTLD following SOT, median overall survival was <3 months from diagnosis.[8]
- There is currently an unmet need for treatment in these patient populations.[5–8]

Tabelecleucel

- Tabelecleucel is an off-the-shelf, allogeneic, T-cell immunotherapy selected for each patient from an existing library based on an appropriate human leukocyte antigen (HLA) restriction and allele profile. The manufacturing process is described in **Figure 1**.
- In EBV+ PTLD patients following SOT or HCT and after failure of rituximab, or rituximab and chemotherapy, treatment with tabelecleucel demonstrated encouraging outcomes.
 - In SOT patients, an objective response rate (ORR) of 50% (95% CI, 23.0-77.0) has been elicited. For patients who responded to tabelecleucel, 2-year overall survival (OS) was 86%.[9]
 - In HCT patients an ORR of 68.6% (95% CI, 50.7-83.1) has been elicited. For patients who responded to tabelecleucel, 2-year OS was 83%.[9]
- Here, we describe the design of an ongoing phase 3 study (ALLELE) assessing the efficacy and safety of tabelecleucel for SOT and HCT patients with EBV+ PTLD after failure of rituximab ± chemotherapy. ALLELE (NCT03394365) has recently been amended to enroll patients with EBV+ PTLD following HCT in addition to SOT (HCT patients formerly in study MATCH [NCT03392142]).

Figure 1. Off-the-shelf, Allogeneic Tabelecleucel Manufacturing Process

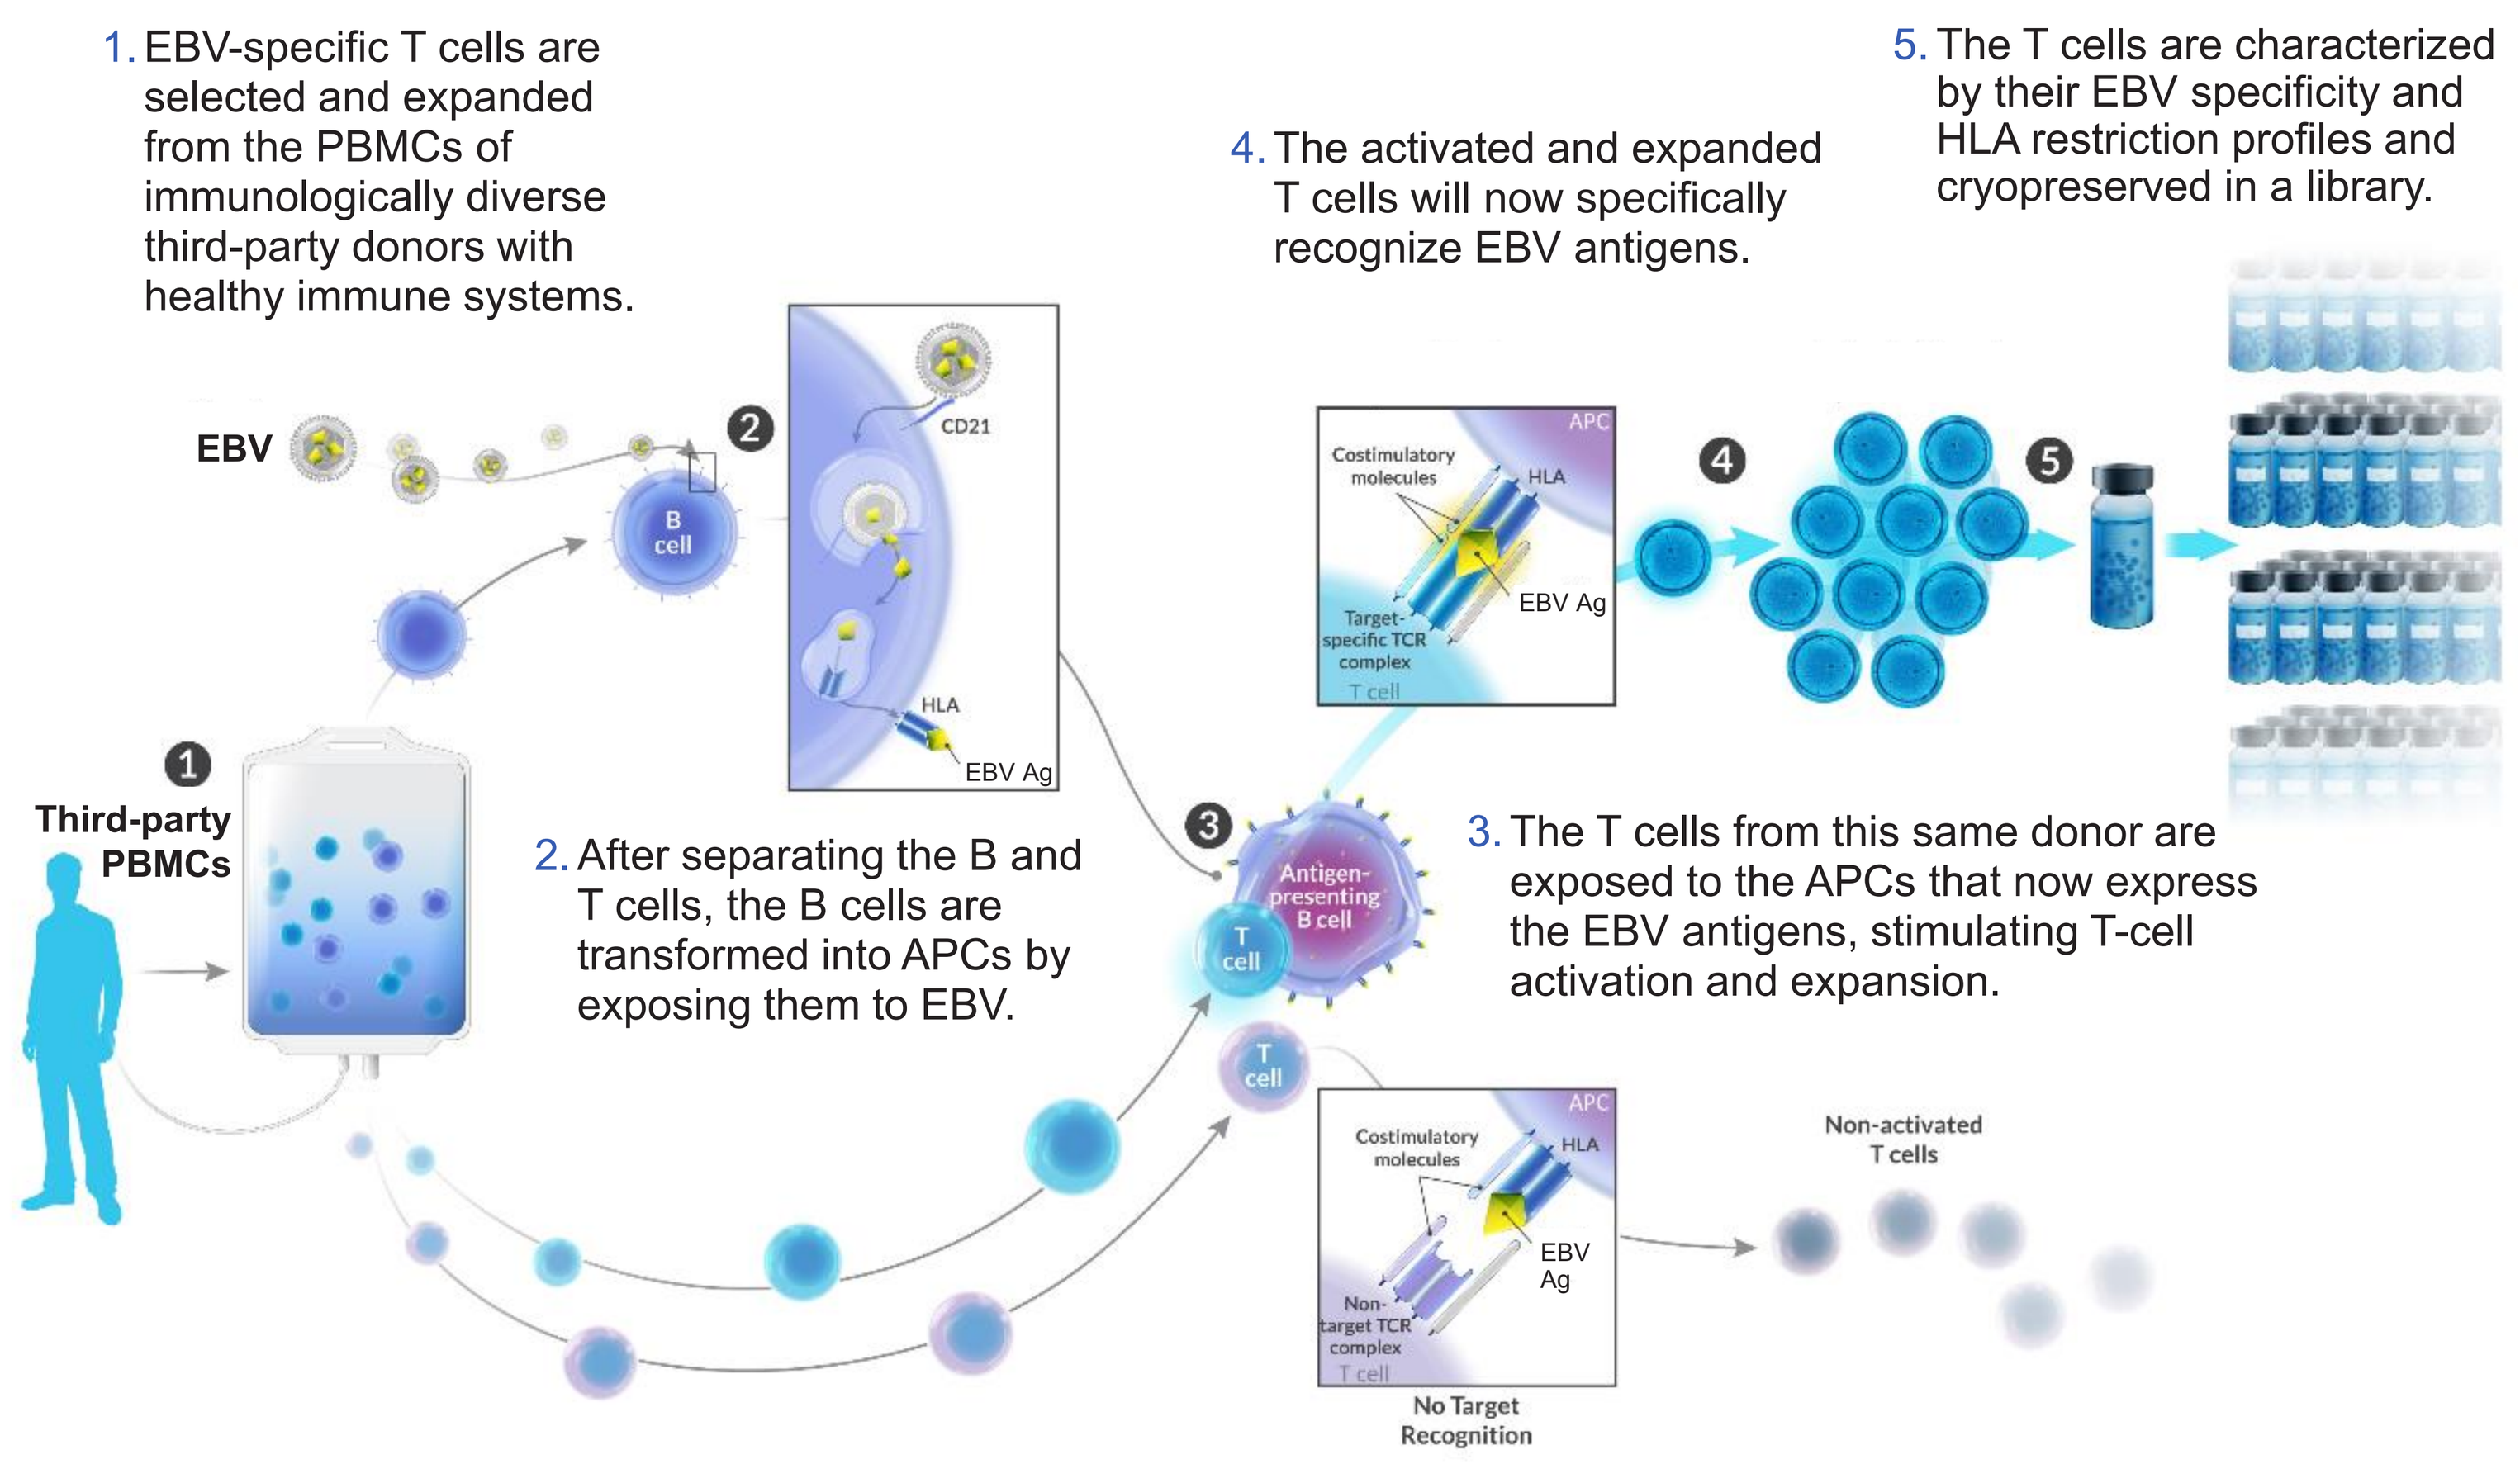
1. EBV-specific T cells are selected and expanded from the PBMCs of immunologically diverse third-party donors with healthy immune systems.
4. The activated and expanded T cells will now specifically recognize EBV antigens.
5. The T cells are characterized by their EBV specificity and HLA restriction profiles and cryopreserved in a library.
EBV
B cell
CD21
HLA
EBV Ag
Costimulatory molecules
APC
Target-specific TCR complex
T cell
Third-party PBMCs
2. After separating the B and T cells, the B cells are transformed into APCs by exposing them to EBV.
Antigen-presenting B cell
3. The T cells from this same donor are exposed to the APCs that now express the EBV antigens, stimulating T-cell activation and expansion.
Non-target TCR complex
Non-activated T cells
No Target Recognition

APC = antigen-presenting cell; HLA = human leukocyte antigen; PBMC = peripheral blood mononuclear cell; TCR = T-cell receptor.

STUDY DESIGN

Figure 2. ALLELE Study Design

Patients receive additional treatment cycles (1 cycle = 35 days) until they meet end of treatment criteria

Tabelecleucel Inventory Check*
availability of tabelecleucel with an appropriate HLA restriction and allele profile

→ **Screening**

→ **HCT Cohort (n=33†)**
Failure of rituximab

→ **SOT Cohort (n=33†)**
Subgroup A: failure of rituximab
Subgroup B: failure of rituximab + chemotherapy

→ **Treatment Cycle (35 days)**
Tabelecleucel is administered intravenously at a dose of 2×10^6 cells/kg on dosing Days 1, 8 and 15
Pre-dose assessment‡

→ **Clinical and Radiographic Assessment at Day ~35**
- Investigator assessments to determine additional treatment cycles using the modified Lugano Classification criteria[10]
- IORA independent review for primary efficacy assessment
- Switching to tabelecleucel with a different HLA restriction will occur after 2 consecutive responses of SD or any assessment of PD during treatment

→ **End of Treatment**
Treatment ends with:
- **Maximal response achieved** (reached when the patient has received 3 consecutive PR or 2 consecutive CR assessments, as assessed by the Lugano Classification response criteria)
- **Unacceptable toxicity** (any grade GvHD [SOT cohort] or grade ≥ 3 GvHD [HCT cohort]; > Grade 3 CRS, any grade 4 non-hematologic AE)
- **Initiation of non-protocol therapy**
- **Failure of tabelecleucel with up to 2 (SOT) or 4 (HCT) different HLA restrictions**

→ **Follow-up**
Disease response assessment every 3 months up to 24 months, and every 6 months thereafter for survival status up to 2 years

*Cytomegalovirus serostatus, DNA based high-resolution HLA typing, and patient's weight and demographics.

†Estimated cohort size.

‡Assessments are performed within 7 days before cycle 2 and thereafter, and include the following: ECOG status/Lansky score, lactate dehydrogenase, serum EBV DNA, radiographic assessments, hematology, chemistry and anti-HLA antibodies.

AE = adverse event;
CR = complete response;
CRS = cytokine release syndrome;
EBV+ PTLD = Epstein-Barr virus post-transplant lymphoproliferative disease;
ECOG = Eastern Cooperative Oncology Group;
GvHD = graft-versus-host disease;
HCT = hematopoietic stem cell transplant;
HLA = human leukocyte antigen;
IORA = independent oncologic response adjudication;
PD = progressive disease;
PR = partial response;
SD = stable disease;
SOT = solid organ transplant.

ALLELE Endpoints

Primary Endpoint

- ORR – complete response or partial response – obtained following administration of tabelecleucel with up to two different HLA restrictions in the SOT or HCT cohort evaluated by independent oncologic response adjudication (IORA).

Secondary Endpoints

- Duration of response (DOR) in SOT and HCT cohorts separately
- ORR and DOR in SOT and HCT cohorts combined
- Rate of complete response and partial response
- Time to response and time to best response
- Overall survival
- Rates of allograft loss/rejection episodes (for SOT cohort only)

ALLELE Inclusion Criteria

- Prior SOT of kidney, liver, heart, lung, pancreas, small bowel, or any combination of these (SOT cohort); or prior allogeneic HCT (HCT cohort)
- A diagnosis of locally assessed, biopsy-proven EBV+ PTLD
- Confirmation of available appropriate partially HLA-matched and restricted tabelecleucel
- Measurable systemic disease using Lugano Classification by PET/CT
- Treatment failure of rituximab or interchangeable commercially available biosimilar monotherapy (HCT and SOT subgroup A) or rituximab plus any concurrent or sequentially administered chemotherapy regimen (SOT subgroup B) for treatment of PTLD. Treatment failure is defined based on rituximab response as follows:
 a. Radiographic disease progression per Lugano Classification following a minimum cumulative dose of 1125 mg/m^2 rituximab (typically, 3 weekly doses of 375 mg/m^2), or
 b. Failure to achieve CR or PR, defined by Lugano radiographic criteria, after a minimum cumulative dose of 1500 mg/m^2 rituximab (typically, 4 weekly doses of 375 mg/m^2), or
 c. Relapse/progression of PTLD after a response to rituximab (SOT subgroup A or HCT cohort) or rituximab plus chemotherapy (SOT subgroup B), defined as radiographic and/or biopsy evidence of relapse/progression consistent with PTLD; if the underlying disease for which the patient underwent allogeneic HCT (HCT cohort) was lymphoma, biopsy confirmation of relapsed EBV+ PTLD is required
- Males and females of any age
- ECOG performance status ≤3 for patients aged >16 years; Lansky score ≥20 for patients from birth to 16 years
- If allogeneic HCT was performed as treatment for an acute lymphoid or myeloid malignancy, the underlying primary disease for which the patient underwent transplant must be in morphologic remission (HCT cohort only)
- Adequate organ function

ALLELE Exclusion Criteria

- Daily steroids of >0.5 mg/kg prednisone or glucocorticoid equivalent, ongoing methotrexate, or extracorporeal photopheresis
- Untreated CNS PTLD, or CNS PTLD for which the patient is actively receiving treatment
- Grade ≥2 graft-versus-host disease
- Burkitt lymphoma, classical Hodgkin lymphoma, or any T cell lymphoma
- Active adenovirus viremia (HCT cohort only)
- Need for vasopressor or ventilatory support
- Antithymocyte globulin or similar anti-T cell antibody therapy ≤4 weeks prior to enrollment
- Treatment with EBV T cells or CAR T cells directed against B cells within 8 weeks of enrollment (SOT or HCT cohorts); or unselected donor lymphocyte infusion within 8 weeks of enrollment (HCT cohort only)

CAR = chimeric antigen receptor; CNS = central nervous system; CR = complete response; CT = computed tomography; EBV = Epstein-Barr virus; EBV+ PTLD = Epstein-Barr virus post-transplant lymphoproliferative disease; ECOG = Eastern Cooperative Oncology Group; HCT = hematopoietic stem cell transplant; HLA = human leukocyte antigen; PET = positron emission tomography; PR = partial response; SOT = solid organ transplant.

Now Enrolling

For more information about the ALLELE clinical study please contact

clinicalstudies@atarabio.com

or visit

https://www.atarabio.com/medical-professionals/

REFERENCES

1. Nijland ML, et al. *Transplant Direct.* 2015;15;2:e48.
2. Ocheni S, et al. *Bone Marrow Transplant.* 2008;42:181–6.
3. Uhlin M, et al. *Haematologica.* 2014;99:346–52.
4. Swinnen LJ, et al. *Transplantation.* 2008;86:215–22.
5. Fox CP, et al. *Bone Marrow Transplant.* 2014;49:280–6.
6. González-Barca E, et al. *Haematologica.* 2007;92:1489–94.
7. Choquet S, et al. *Haematologica.* 2007;92:273–4.
8. Zimmermann H, et al. EHA Library. 2019;266518;PF719.
9. Prockop S, et al. EHA Library. 2018;214869;PF401 and associated poster presentation
10. Cheson BD, et al. *J Clin Oncol.* 2014;32:3059–68.

DISCLOSURES AND ACKNOWLEDGMENTS

This clinical study is funded by Atara Biotherapeutics. Susan Prockop is a co-inventor on intellectual property licensed to Atara. Dr. Prockop has waived her rights to this intellectual property to MSK, and has no personal financial interests in Atara. Dr. Prockop does receive research funding in support of sponsored clinical trials from Atara through MSK. MSK has financial interests in Atara and intellectual property interests relevant to the work that is related to this abstract. Minoti Hiremath, Wei Ye, Laurence Gamelin and Willis H. Navarro are employees of Atara. Kris Michael Mahadeo receives consultancy fees from Atara.

Medical writing assistance was provided by Anna Sanniti, PhD from Seques (AMICULUM Ltd), funded by Atara.

Transplantation and Cellular Therapy Meeting of ASTCT and CIBMTR, 2020, Orlando. February 19–23, 2020